Abstract 2554: Placental Circulating T Cells Expressing CD16 Demonstrate Potent Efficacy Against Multiple Hematological and Solid Tumor Cancers Through Combination with Various Monoclonal Antibodies

Kathy Karasiewicz-Mendez, Kristina Tess, Mansour Djedaini, Natalia Ruggeri Barbaro, Chenfei Huang, Joseph Gleason, Shengchen Lin, John Fitzgerald, Shuyang He, Robert Hariri, and Adrian Kilcoyne

Celularity Inc., Florham Park, New Jersey.

Background

- Celularity Inc. is developing PT-CD16VS as a novel platform that can be combined with various monoclonal antibodies to steer antigen recognition and engage in antibody-dependent cell cytotoxicity (ADCC) against diverse cancer types with a "Universal receptor/product" approach. PT-CD16VS is an allogeneic cell therapy product derived from human postpartum placental circulating T (P-T) cells that are genetically modified to express a proprietary CD16 variant with knockout of the endogenous T cell receptor (TCR-α/β).
- Here we report preclinical evaluation of PT-CD16VS in combination with various monoclonal antibodies against tumor cell lines originating from both solid tumors and hematological cancers.

Methods

- PT-CD16VS Drug Product Manufacturing & Characterization: PT-CD16VS cells were generated through transduction of human placental circulating T cells using a lentiviral vector containing a CD16 construct expressing a high affinity CD16 variant (CD16VS) followed by knock-out of the TCR through transfection. The phenotype of PT-CD16VS cells was determined using flow cytometry. The cells were stained for CD56, CD20, CD5, CD16, and TCR-α/β expression. The viability was assessed using 7AAD staining. For targets, cells were stained for HER2, PD-L1, EGFR, or CD20 expression.
- Cytotoxicity Assay: *In vitro,* the ADCC activity of PT-CD16VS cells against solid tumors and hematological cancers was assessed using a kinetic ACEA–based cytotoxicity assay. The functional activity of PT-CD16VS cells was tested in combination with various monoclonal antibodies: Trastuzumab (1µg/mL) against HER2^{+} solid tumors, Avelumab (0.1µg/mL) against PD-L1^{+} solid tumors, Cetuximab (0.1µg/mL) against EGFR^{+} solid tumors, and Rituximab (1µg/mL) against CD20^{+} hematological cancers.
- Cytokine Release Assay: *In vitro,* the functional activity of PT-CD16VS cells against solid tumors and hematological cancers was assessed using a Cytokine Release Assay. PT-CD16VS cells along with different monoclonal antibodies (same concentrations as in ADCC) were co-cultured with target cells at an Effector to Target (E:T) ratio of 1:1 for 24 hours, followed by cytokine quantification using Meso Scale Discovery (MSD) platform (for JIMT-1, NCI-H1975, NCI-N87, and Daudi) and Luminex platform (for MDA-MB-231 and NSALEC).
- Proliferation Assay: *In vitro,* PT-CD16VS proliferation in combination with Trastuzumab (1µg/mL) against NCI-N87 or with Rituximab (1µg/mL) against Raji was assessed by co-culturing PT-CD16VS cells at an E:T ratio of 1:1. Co-cultures were harvested after 48 and 72 hours (NCI-N87) and 5 days (Raji) of culture, counted, and analyzed by flow cytometry to enumerate the total number of PT-CD16VS cell per co-culture.
- *In-vivo* Anti-Tumor Model: PT-CD16VS with 10mg/kg Trastuzumab was evaluated in a subcutaneous NCI-N87 xenograft model in NSG mice. Additionally, PT-CD16VS was combined with 2mg/kg Rituximab in a disseminated Raji-luciferase xenograft model in NSG mice.
- Cancer artwork shown in figures were obtained/ adapted from pictures provided by Servier Medical Art (Servier; https://smart.servier.com/), licensed under a Creative Commons Attribution 4.0 Unported License.

Results

Figure 1. PT-CD16VS Drug Product Exhibited High Viability, T Cell Purity, and CD16 Transduction Efficiency

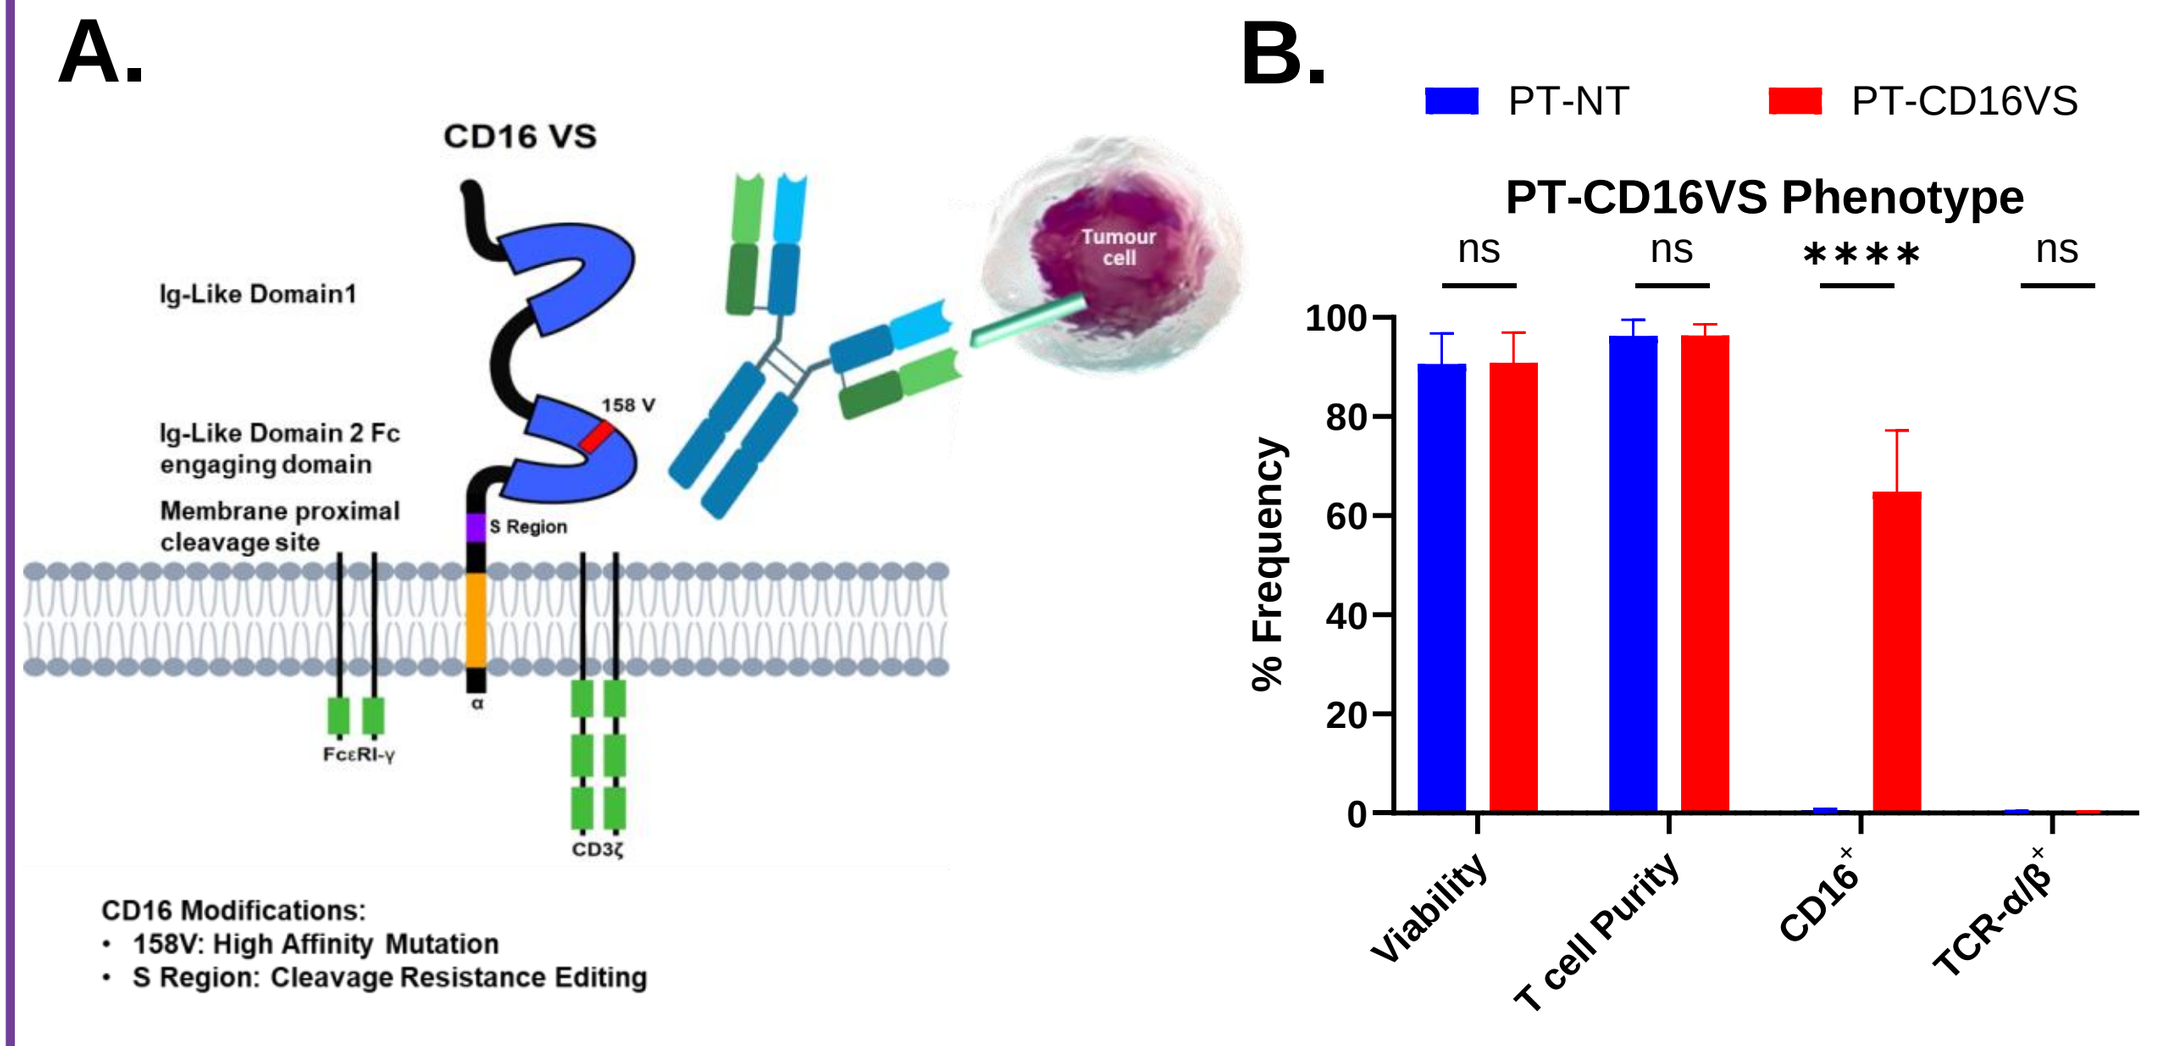
A.
CD16 VS
Ig-Like Domain1
Ig-Like Domain 2 Fc engaging domain
Membrane proximal cleavage site
158 V
S Region
FcεRI-γ
CD3ζ
Tumour cell
CD16 Modifications:
• 158V: High Affinity Mutation
• S Region: Cleavage Resistance Editing
B.
PT-NT
PT-CD16VS
PT-CD16VS Phenotype
% Frequency
Viability
T cell Purity
CD16+
TCR-α/β+

Figure 1. (A) CD16VS construct design. (B) Phenotype of PT-CD16VS and PT Non-transduced (NT) cells (Mean with SD, each group n=11). Statistical analysis performed with multiple paired *t* tests (****p<0.0001).

Figure 2. PT-CD16VS Cells in Combination with Trastuzumab Exhibit Potent Activity Against HER2 Expressing Tumor Cells, but Not Against HER2 Expressing Normal Cells, and Superior *In-vivo* Efficacy in NCI-N87 Subcutaneous Model in NSG Mice

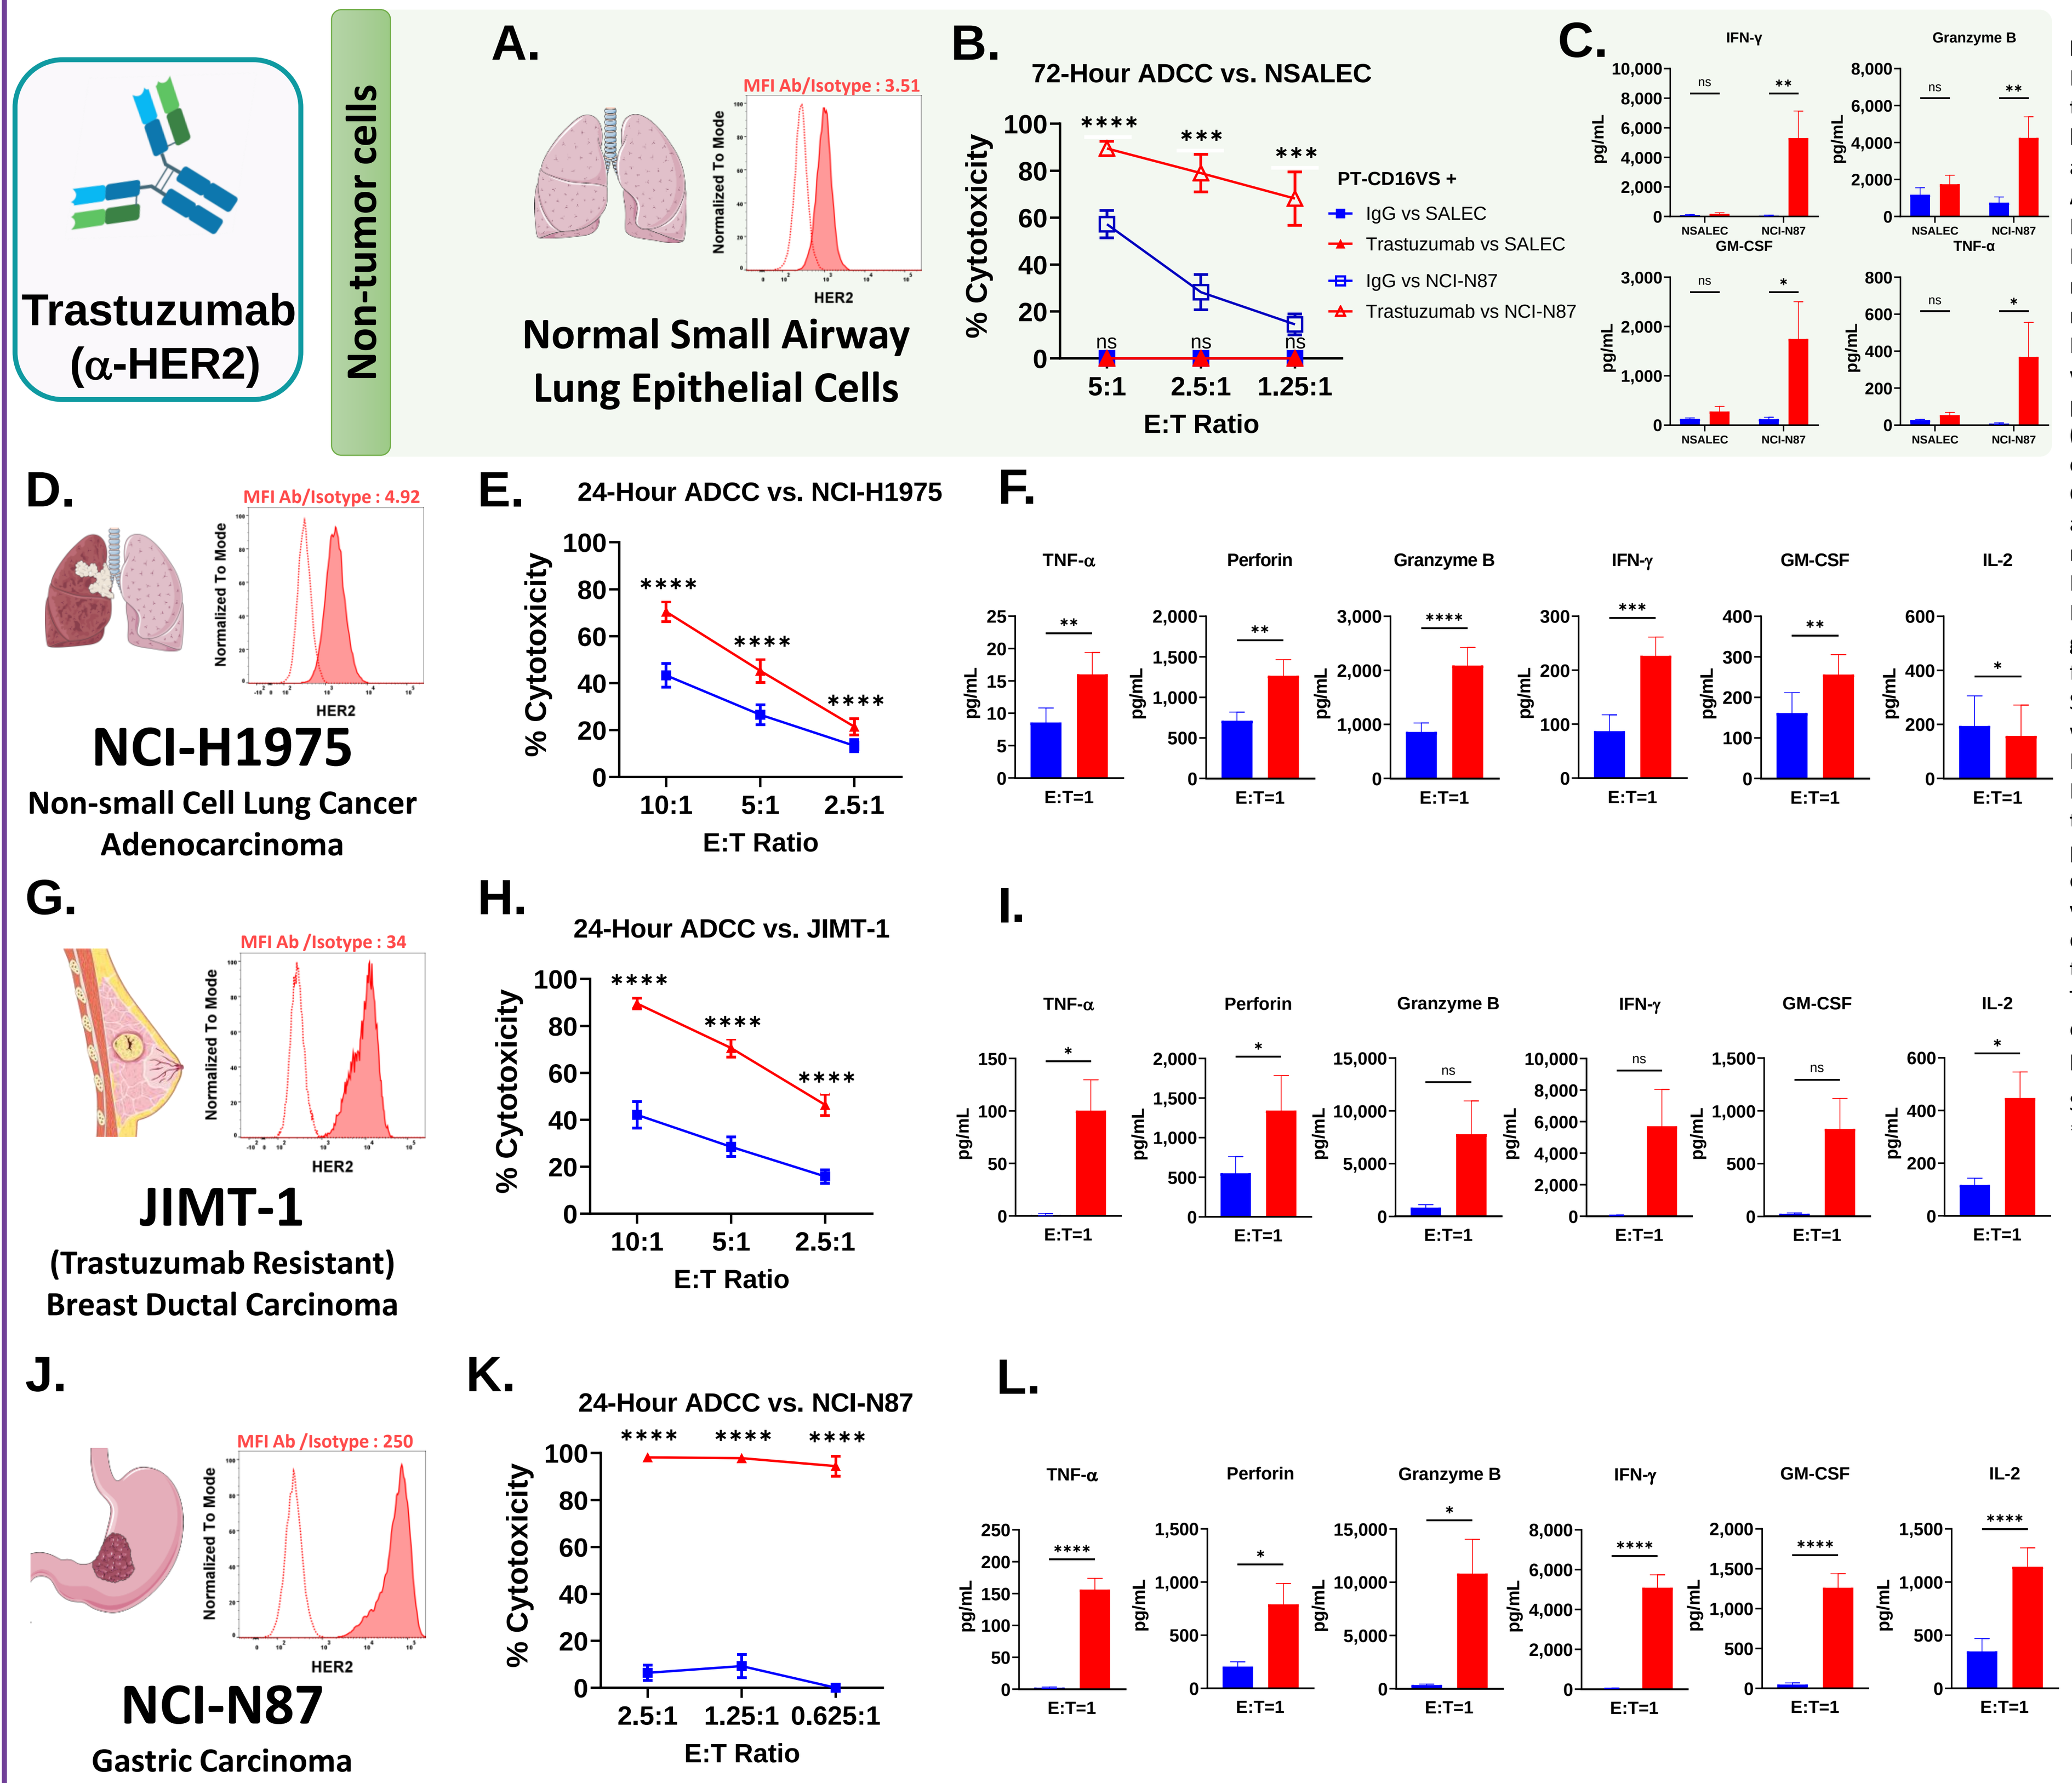
Trastuzumab (α-HER2)
Non-tumor cells
A. Normal Small Airway Lung Epithelial Cells
MFI Ab/Isotype : 3.51
B. 72-Hour ADCC vs. NSALEC
PT-CD16VS +
IgG vs SALEC
Trastuzumab vs SALEC
IgG vs NCI-N87
Trastuzumab vs NCI-N87
C.
D. NCI-H1975
Non-small Cell Lung Cancer Adenocarcinoma
MFI Ab/Isotype : 4.92
E. 24-Hour ADCC vs. NCI-H1975
F.
G. JIMT-1
(Trastuzumab Resistant) Breast Ductal Carcinoma
MFI Ab /Isotype : 34
H. 24-Hour ADCC vs. JIMT-1
I.
J. NCI-N87
Gastric Carcinoma
MFI Ab /Isotype : 250
K. 24-Hour ADCC vs. NCI-N87
L.

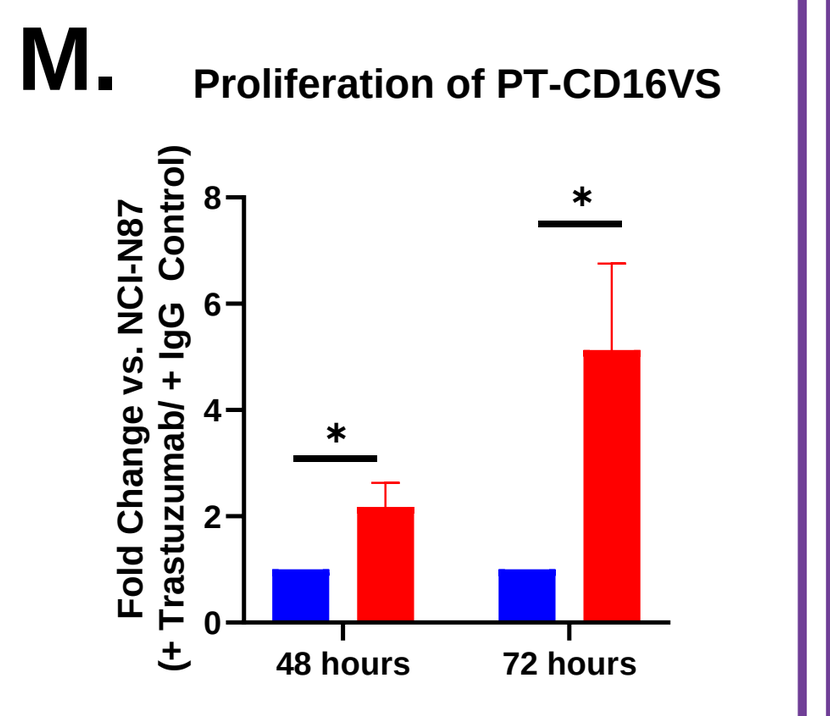
M. Proliferation of PT-CD16VS
48 hours
72 hours

Figure 2. (A),(D),(G),(J) HER2 expression in NSALEC, NCI-H1975, JIMT-1 and NCI-N87 tumor cells in red labeled with PE anti-human HER2 antibody. Isotype- stained cells are shown in dotted line. **(B),(E),(H),(K)** ACEA kinetic cytotoxicity assay against NSALEC (Mean ± SEM, each E:T Ratio n=10), NCI-H1975 (Mean ± SEM, each E:T Ratio n=18), JIMT-1 (Mean ± SEM, each E:T Ratio n=10) and NCI-N87 (Mean ± SEM, each E:T Ratio n=8-11) target cells in combination with Trastuzumab. Statistical analyses performed with multiple paired *t* test. **(C),(F),(I),(L)** Secretion of proinflammatory cytokines and cytolytic proteins by PT-CD16VS in combination with Trastuzumab against NSALEC (Mean ± SEM, each E:T Ratio n=4), NCI-H1975 (Mean ± SEM, each E:T Ratio n=11), JIMT-1 (Mean ± SEM, each E:T Ratio n=4) NCI-N87 (Mean ± SEM, each group n=4-9) tumor cells. Statistical analyses for F, I, L performed with paired *t* test. Statistical analyses for C performed with 2-way ANOVA. **(M)** Proliferation fold change of PT-CD16VS with Trastuzumab against NCI-N87 target cells (Mean ± SEM, each timepoint n=5-7). Statistical analyses performed with unpaired *t* test. **(N)** Schema of NCI-N87 *in-vivo* tumor model. **(O)** Tumor volumes and objective response rates out to day 35 demonstrating superior reduction in tumor volume in the PT-CD16VS + Trastuzumab treated group (Mean ± SEM, each group n=12, Statistical analysis performed using Wilcoxon Rank Sum test).

Statistical analyses: *p<0.05, **p<0.01, ***p<0.005, and ****p<0.0001

PT-CD16VS + IgG
PT-CD16VS + Trastuzumab

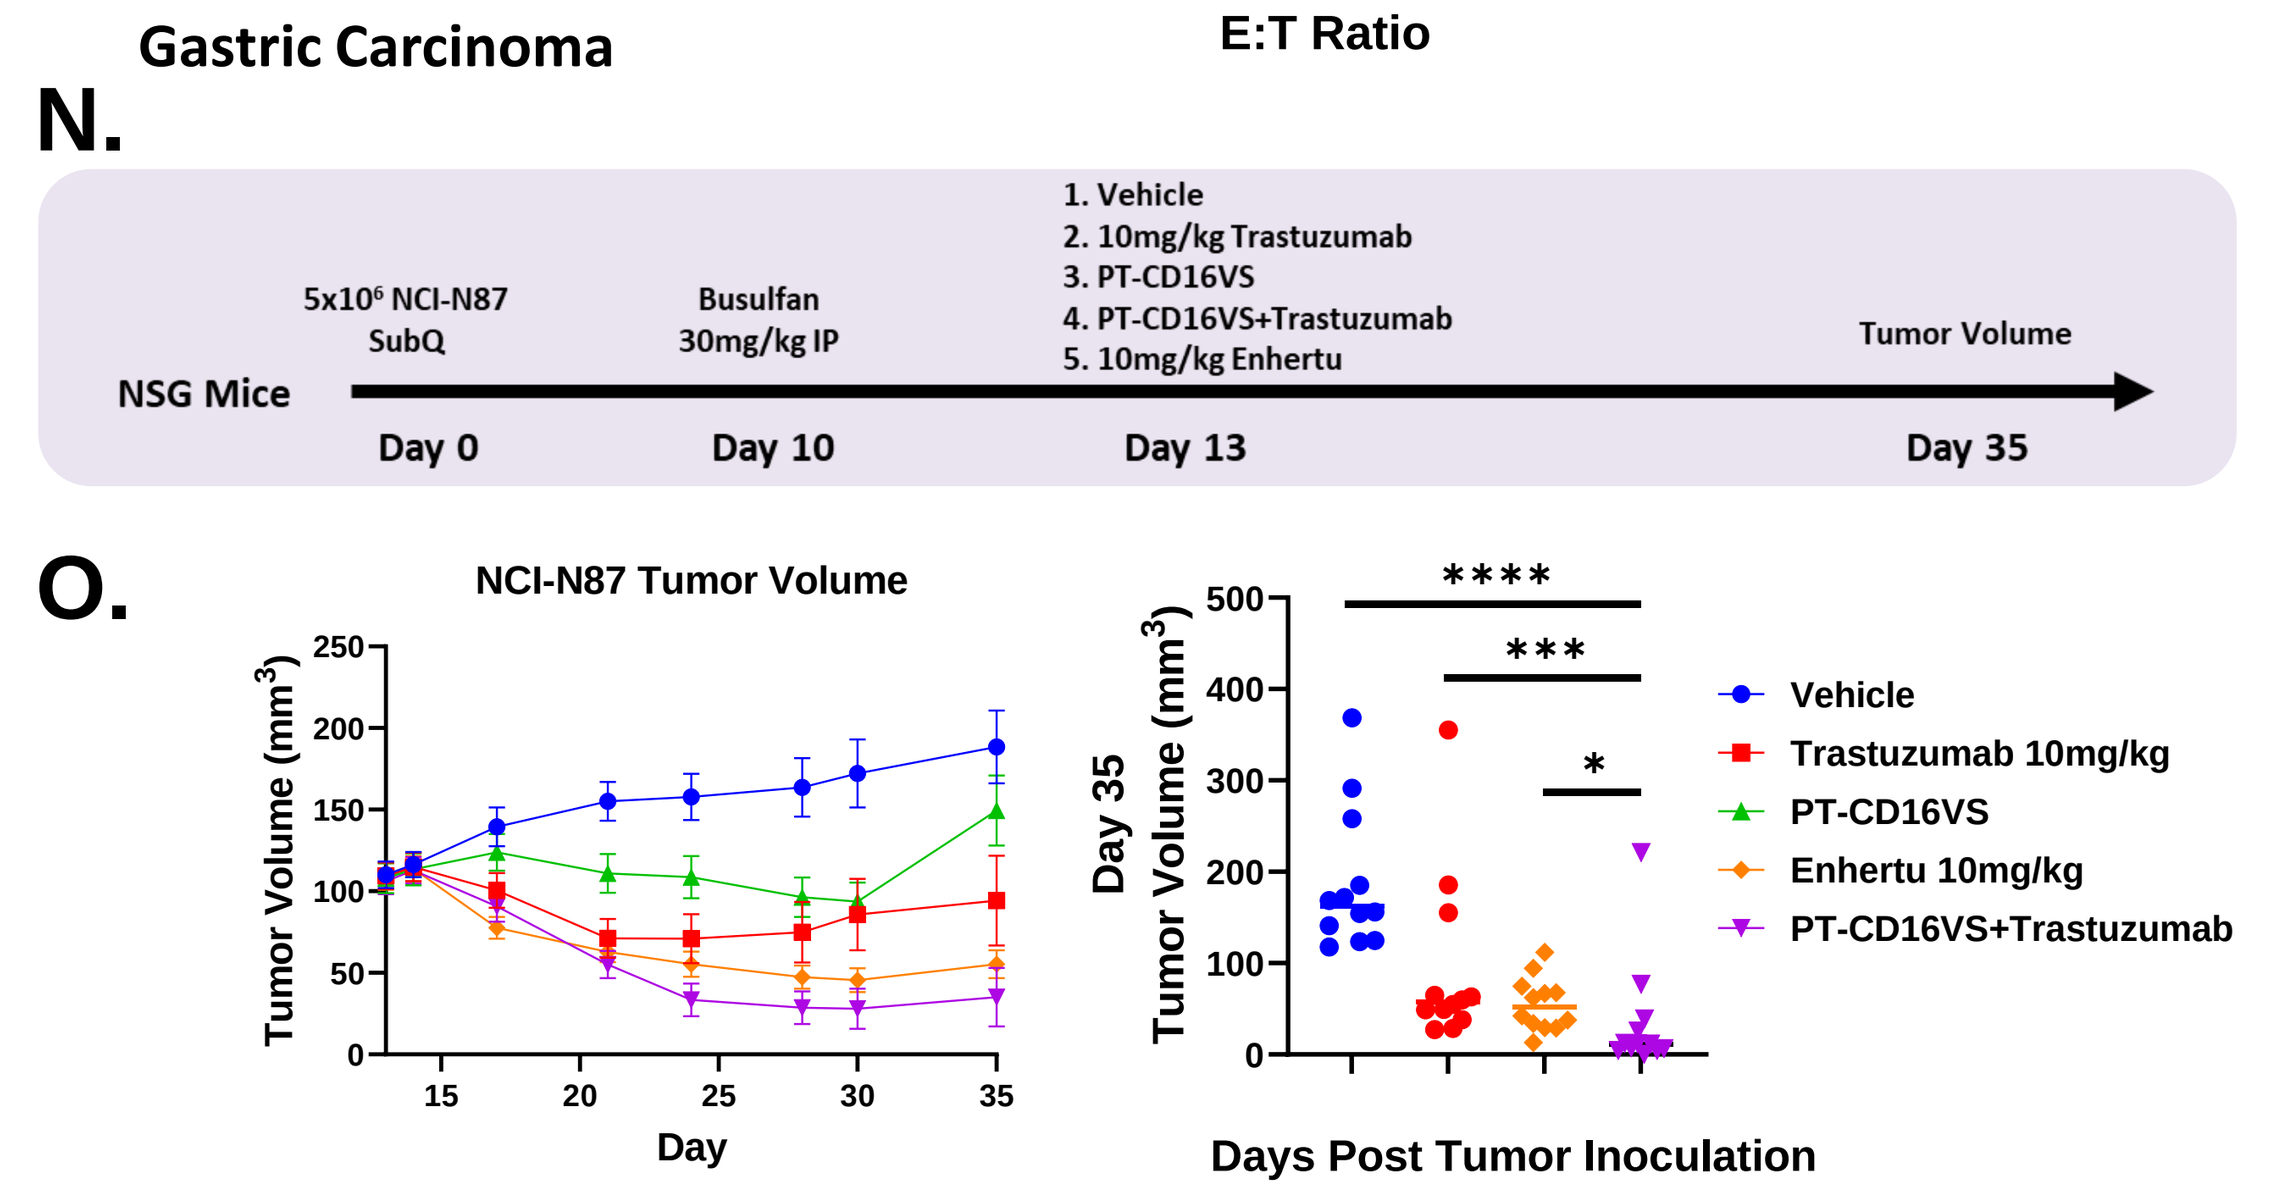
N.
5x10⁶ NCI-N87 SubQ
Busulfan 30mg/kg IP
1. Vehicle
2. 10mg/kg Trastuzumab
3. PT-CD16VS
4. PT-CD16VS+Trastuzumab
5. 10mg/kg Enhertu
Tumor Volume
NSG Mice
Day 0
Day 10
Day 13
Day 35
O.
NCI-N87 Tumor Volume
Tumor Volume (mm³)
Day
Day 35 Tumor Volume (mm³)
Days Post Tumor Inoculation
Vehicle
Trastuzumab 10mg/kg
PT-CD16VS
Enhertu 10mg/kg
PT-CD16VS+Trastuzumab

Day 35 Response			
Group	Complete Response	Partial Response	
		>85% Reduction	30-85% Reduction
Trastuzumab (n=12)	0	0	8/12 (66.7%)
PT-CD16VS+Trast (n=12)	1/12 (8.3%)	7/12 (58.3%)	3/12 (25%)
Enhertu (n=12)	0	1/12 (8.3%)	9/12 (75%)

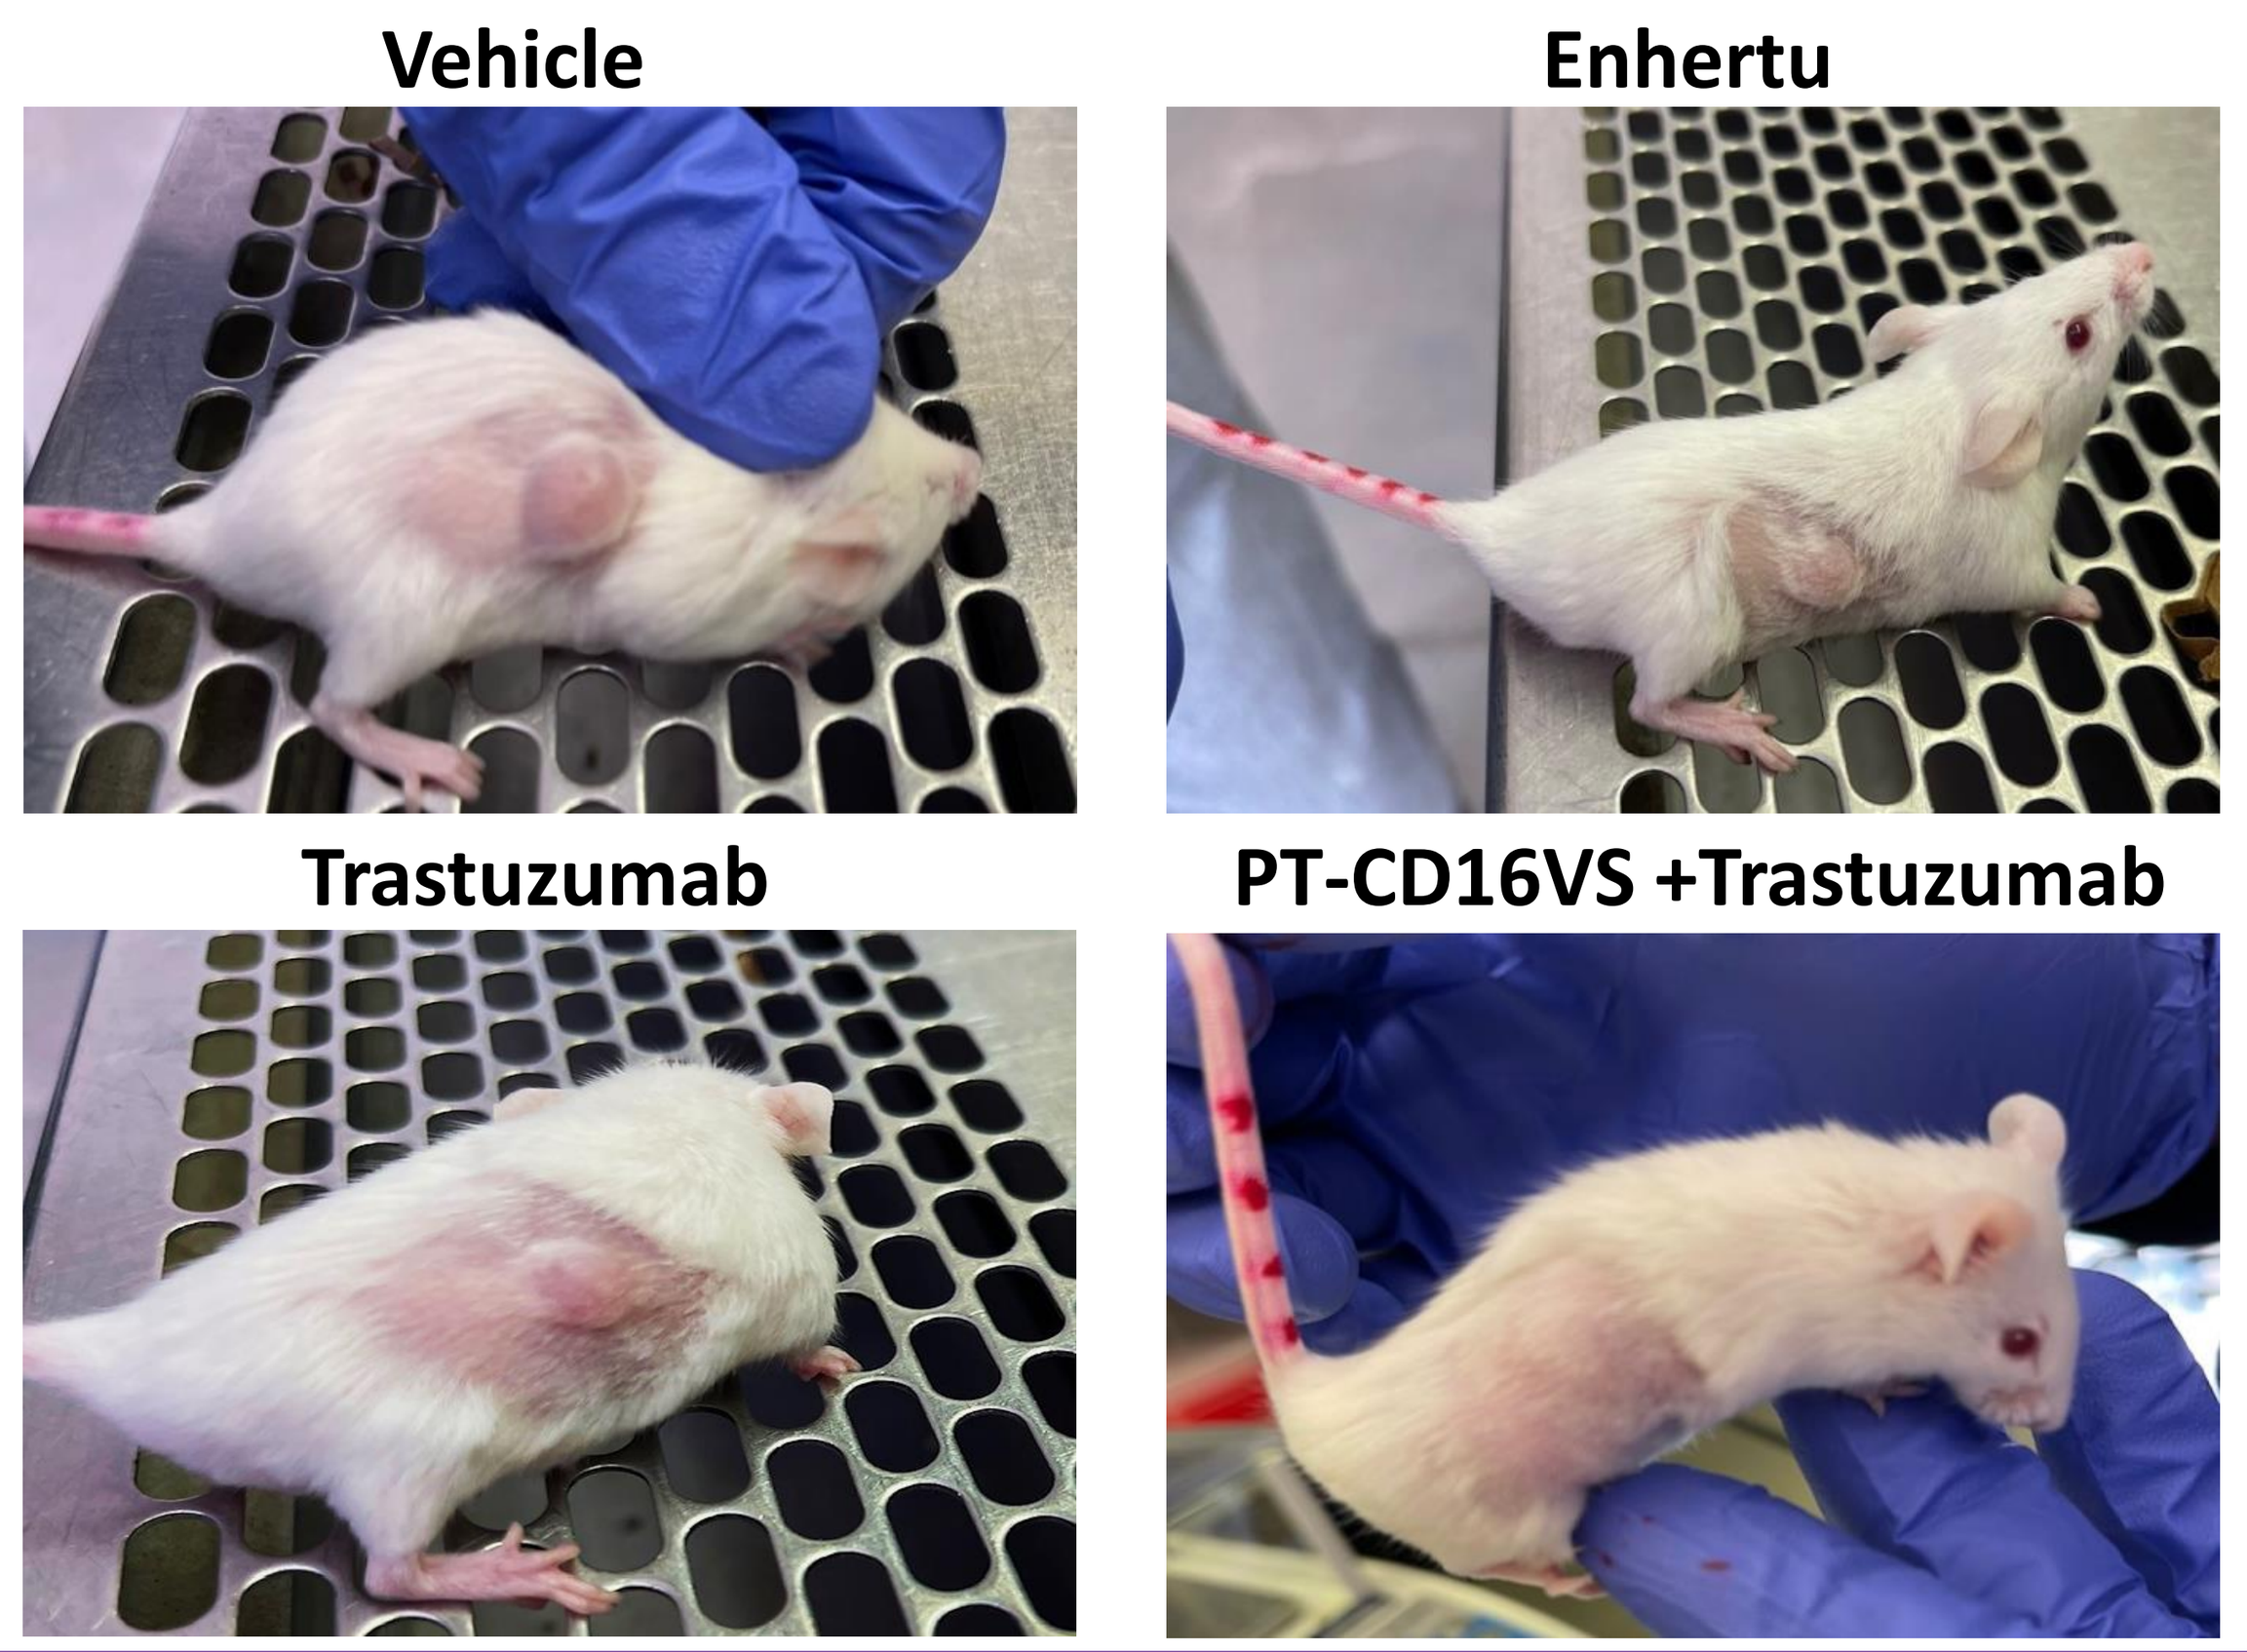
Vehicle
Enhertu
Trastuzumab
PT-CD16VS +Trastuzumab

Figure 3. PT-CD16VS Cells in Combination with Avelumab Exhibit Potent Activity Against PD-L1 Expressing Tumor Cells

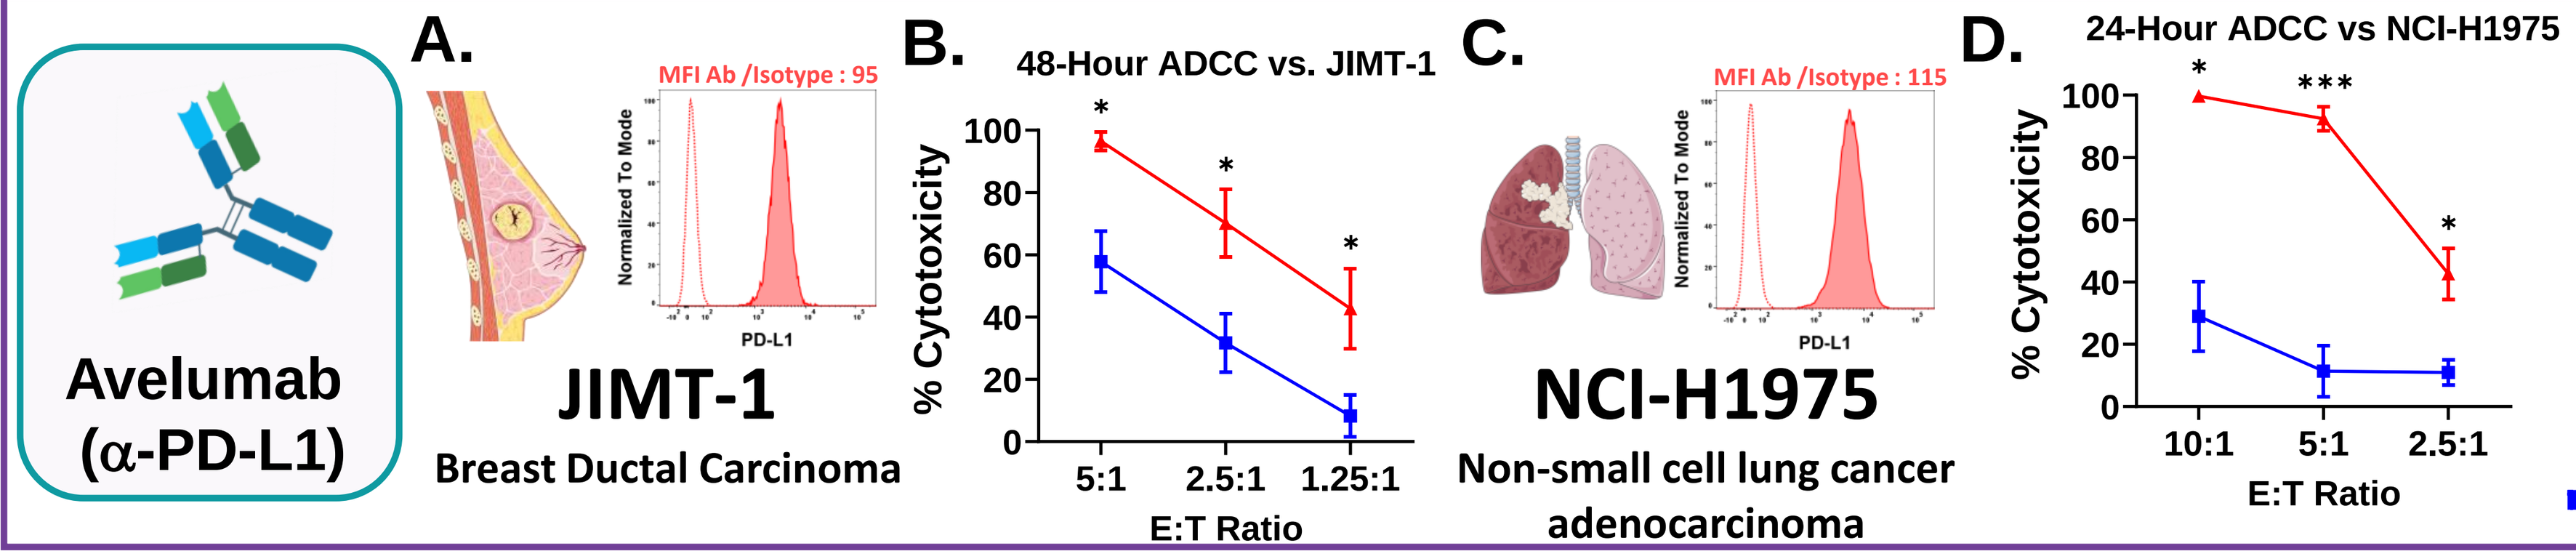
Avelumab (α-PD-L1)
A. JIMT-1
Breast Ductal Carcinoma
MFI Ab /Isotype : 95
B. 48-Hour ADCC vs. JIMT-1
C. NCI-H1975
Non-small cell lung cancer adenocarcinoma
MFI Ab /Isotype : 115
D. 24-Hour ADCC vs NCI-H1975

Figure 3. **(A), (C)** PD-L1 expression in JIMT-1 and NCI-H1975 cells in red labeled with BV785 anti-human PD-L1 antibody. Isotype- stained cells are shown in dotted line. **(B)** ACEA kinetic cytotoxicity assay against JIMT-1 target cells in combination with Avelumab (Mean ± SEM, each E:T Ratio n=3). **(D)** ACEA kinetic cytotoxicity assay against NCI-H1975 target cells in combination with Avelumab (Mean ± SEM, each E:T Ratio n=3). Statistical analyses for B,D performed with paired *t* test (*p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001).

PT-CD16VS + IgG
PT-CD16VS + Avelumab

Figure 4. PT-CD16VS Cells in Combination with Cetuximab Exhibit Potent Activity Against EGFR Expressing Tumor Cells

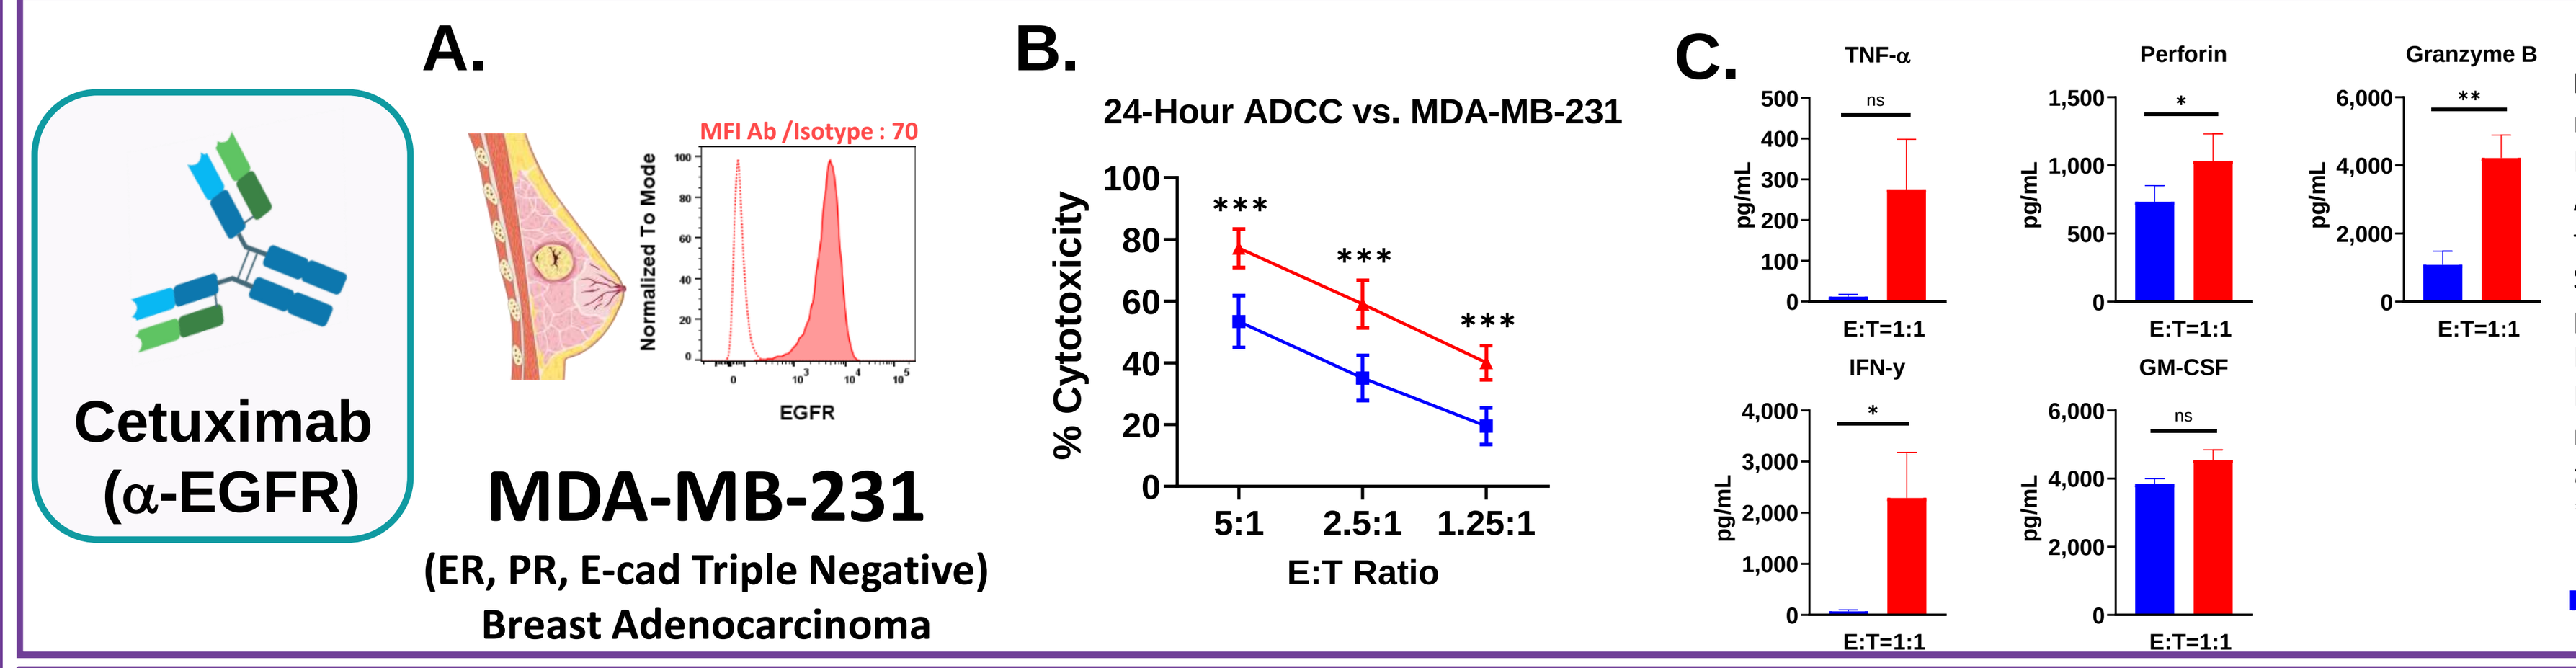
Cetuximab (α-EGFR)
A. MDA-MB-231
(ER, PR, E-cad Triple Negative) Breast Adenocarcinoma
MFI Ab /Isotype : 70
B. 24-Hour ADCC vs. MDA-MB-231
C.

Figure 4. **(A)** EGFR expression in MDA-MB-231 cells in red labeled with APC anti-human EGFR antibody. Isotype- stained cells are shown in dotted line. **(B)** ACEA kinetic cytotoxicity assay against MDA-MB-231 target cells in combination with Cetuximab (Mean ± SEM, each E:T Ratio n=4). **(C)** Secretion of proinflammatory cytokines and cytolytic proteins by PT-CD16VS in combination with Cetuximab against MDA-MB-231 tumor cells. (Mean ± SEM, each group n=4). Statistical analyses for B,C performed with paired *t* test (*p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001).

PT-CD16VS + IgG
PT-CD16VS + Cetuximab

Figure 5. PT-CD16VS Cells in Combination with Rituximab Exhibit Potent Activity Against CD20 Expressing Cancer Cells with Superior *In-vivo* Efficacy in Raji Model in NSG Mice

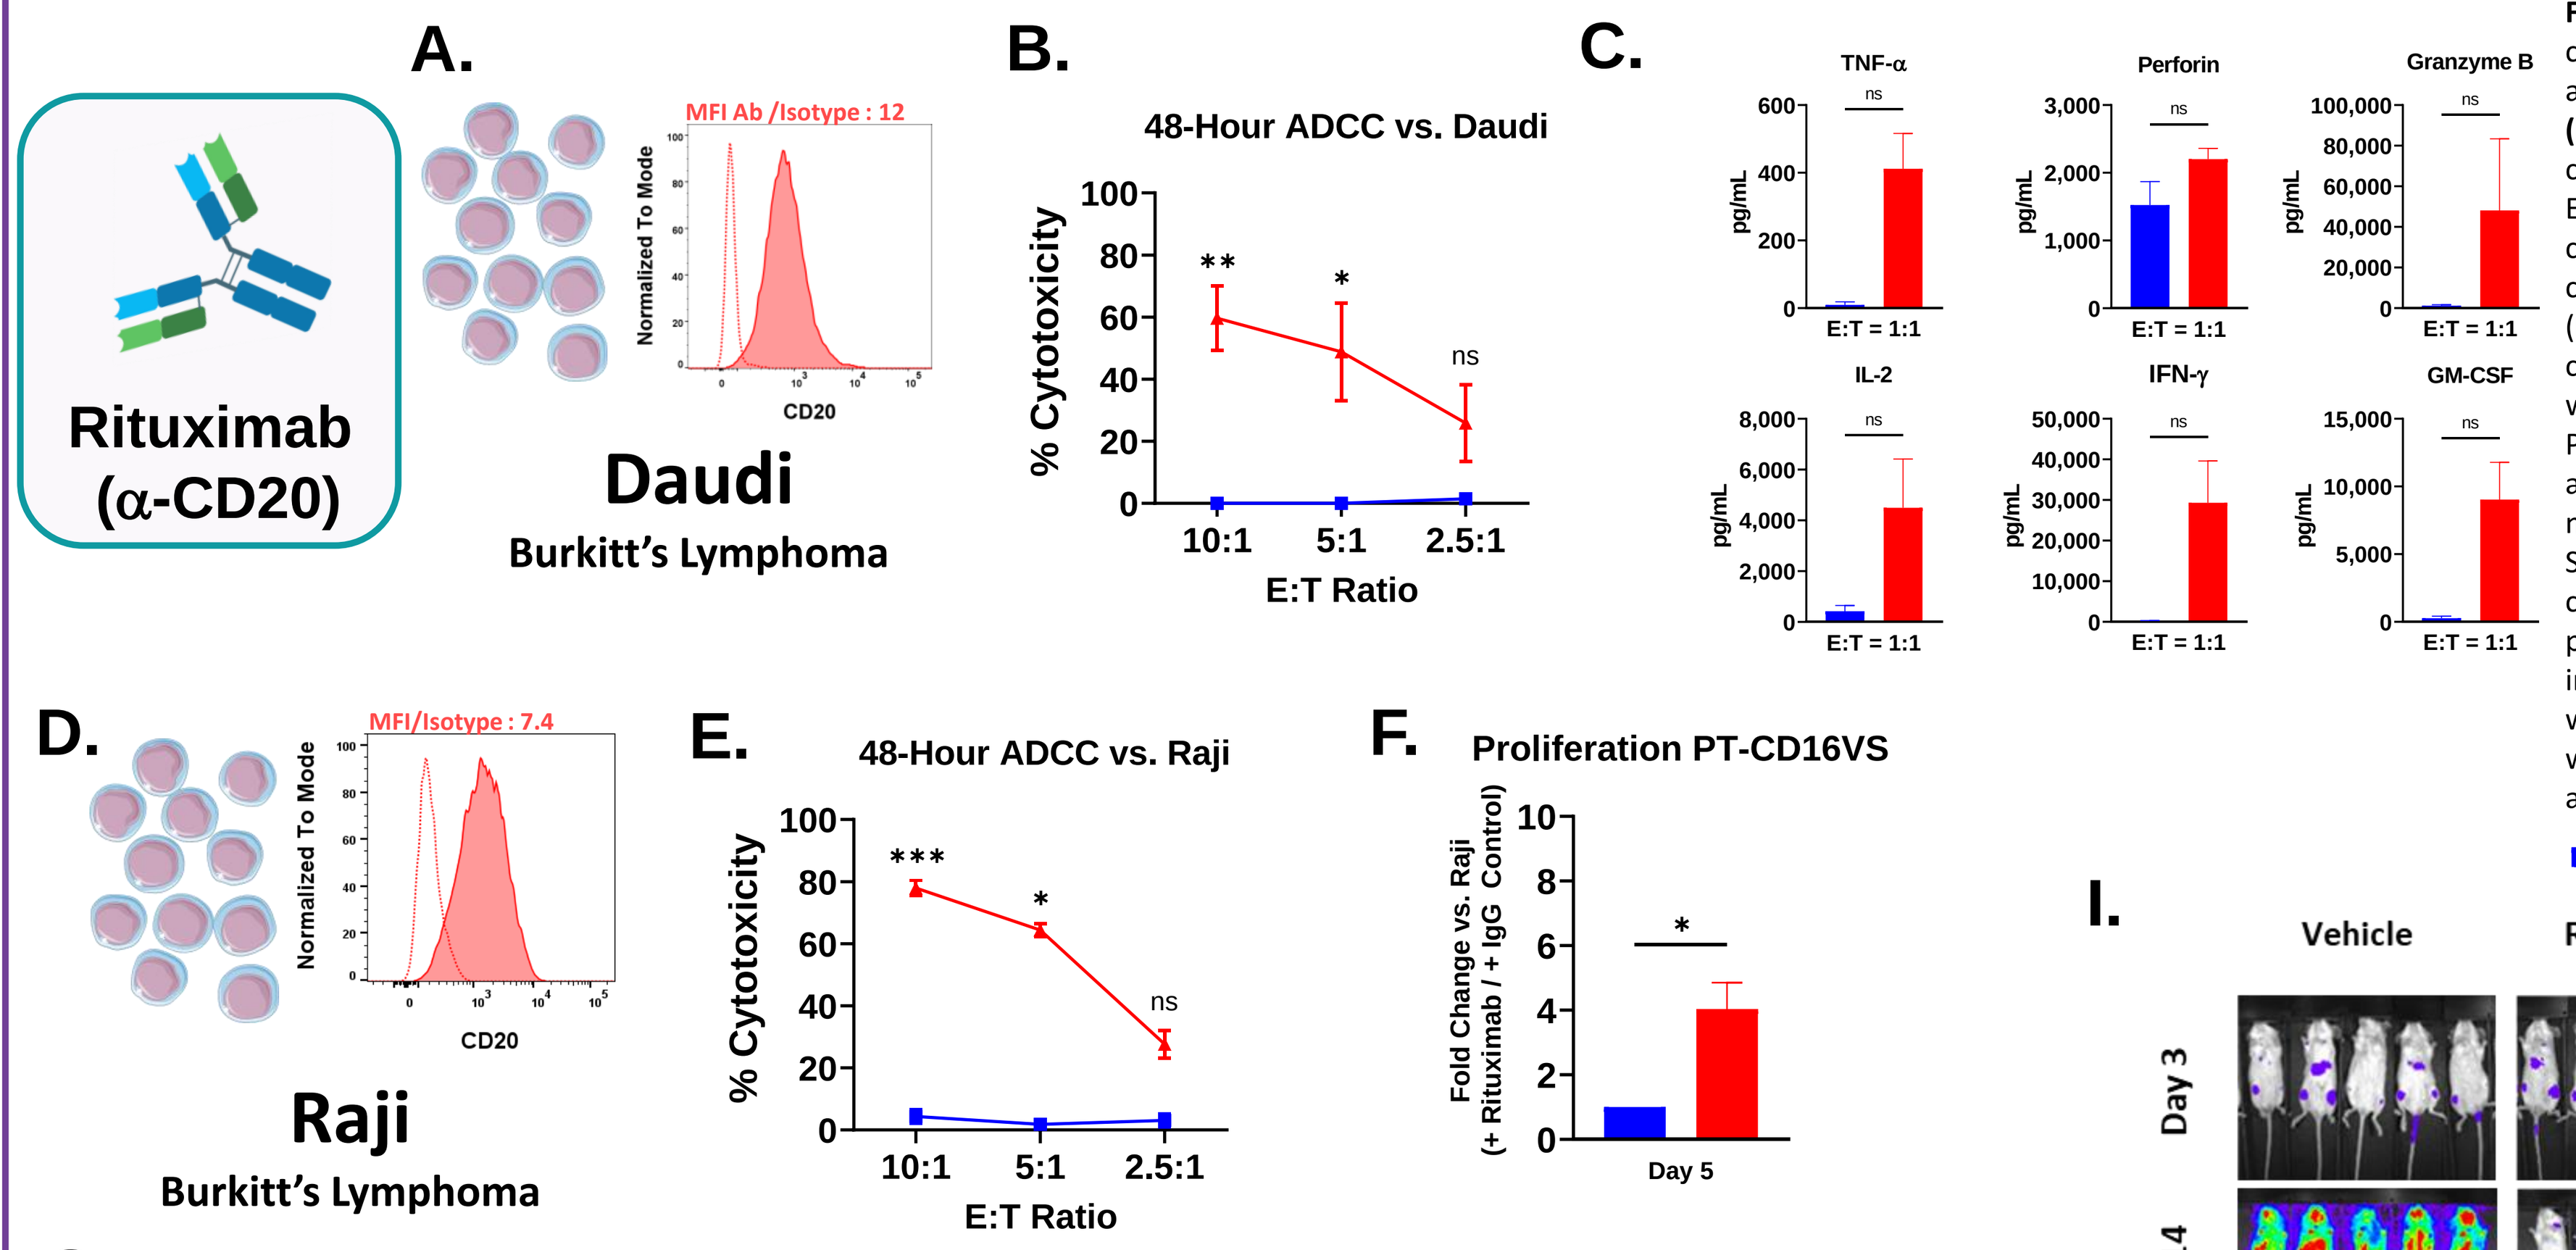
Rituximab (α-CD20)
A. Daudi
Burkitt's Lymphoma
MFI Ab /Isotype : 12
B. 48-Hour ADCC vs. Daudi
C.
D. Raji
Burkitt's Lymphoma
MFI/Isotype : 7.4
E. 48-Hour ADCC vs. Raji
F. Proliferation PT-CD16VS
Day 5

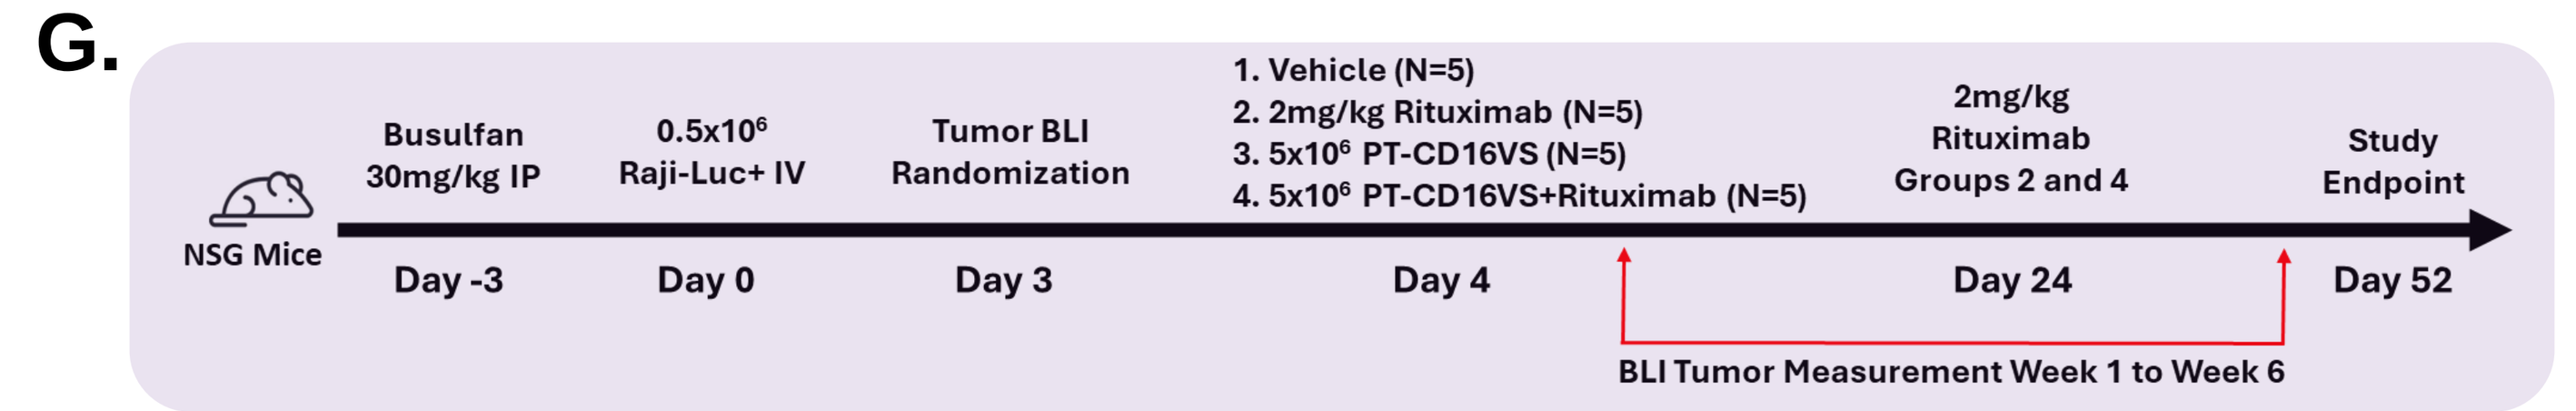
G.
NSG Mice
Busulfan 30mg/kg IP
0.5x10⁶ Raji-Luc+ IV
Tumor BLI Randomization
1. Vehicle (N=5)
2. 2mg/kg Rituximab (N=5)
3. 5x10⁶ PT-CD16VS (N=5)
4. 5x10⁶ PT-CD16VS+Rituximab (N=5)
2mg/kg Rituximab Groups 2 and 4
Study Endpoint
Day -3
Day 0
Day 3
Day 4
Day 24
Day 52
BLI Tumor Measurement Week 1 to Week 6

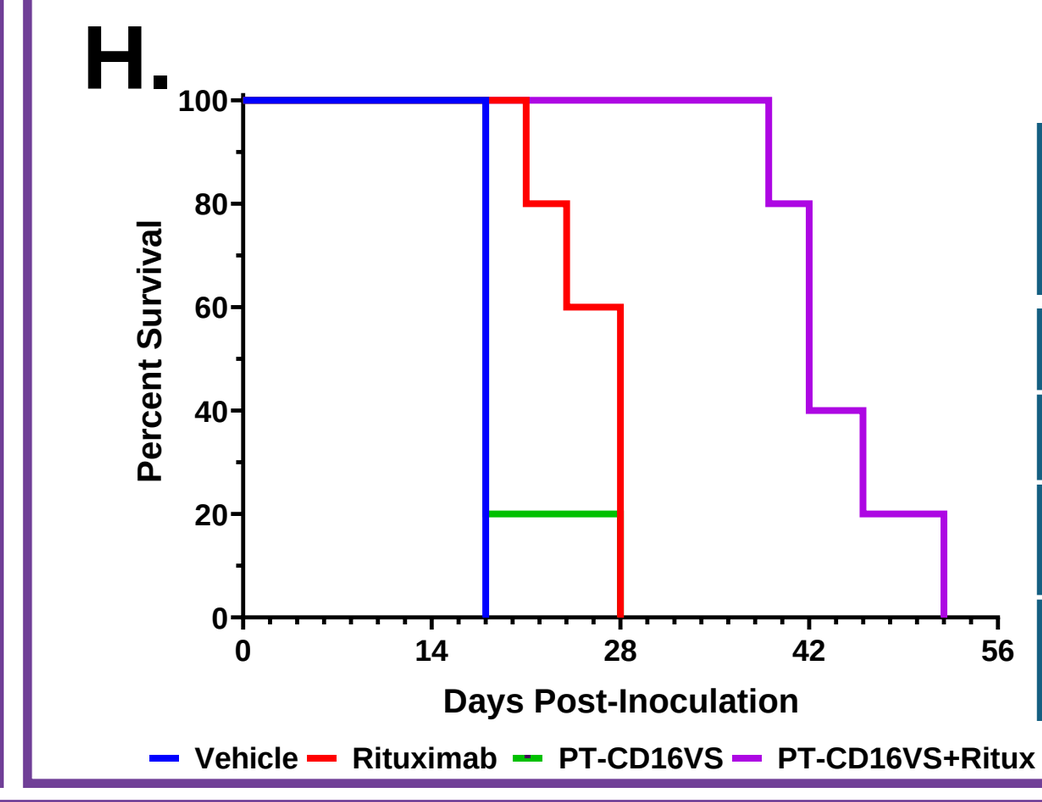
H.
Percent Survival
Days Post-Inoculation
Vehicle
Rituximab
PT-CD16VS
PT-CD16VS+Ritux

Group	Treatment	N	Median Survival	P Values		
				Vs. Group 1	Vs. Group 2	Vs. Group 4
1	Vehicle	5	18	-	0.0027	0.0027
2	Rituximab	5	28	0.0027	-	0.0034
3	PT-CD16VS	5	18	0.3173	0.0965	0.0016
4	PT-CD16VS +Rituximab	5	42	0.0027	0.0034	-

Figure 5. (A), (D) CD20 expression in Daudi or Raji cancer cells in red labeled with BV605 anti-human CD20 antibody. Isotype- stained cells are shown in dotted line. **(B)** ACEA kinetic cytotoxicity assay against Daudi target cells in combination with Rituximab (Mean ± SEM, each E:T Ratio n=3). **(C)** Secretion of proinflammatory cytokines and cytolytic proteins by PT-CD16VS in combination with Rituximab against Daudi cancer cells (Mean ± SEM, each group n=3). **(E)** ACEA kinetic cytotoxicity assay against Raji target cells in combination with Rituximab (Mean ± SEM, each E:T Ratio n=3). **(F)** Proliferation fold change of PT-CD16VS with Rituximab against Raji target cells (Mean ± SEM, each timepoint n=3). **(G)** Schema of *in-vivo* Raji tumor model. **(H)** Survival curve analysis of PT-CD16VS treated mice in combination with Rituximab. Statistical analysis performed using Log-Rank (Mantel-Cox) test. **(I)** BLI imaging of mice. Statistical analyses for B,C,E performed with paired *t* test, statistical analyses for F performed with unpaired *t* test (*p<0.05, **p<0.01, ***p<0.001 and ****p<0.0001).

PT-CD16VS + IgG
PT-CD16VS + Rituximab

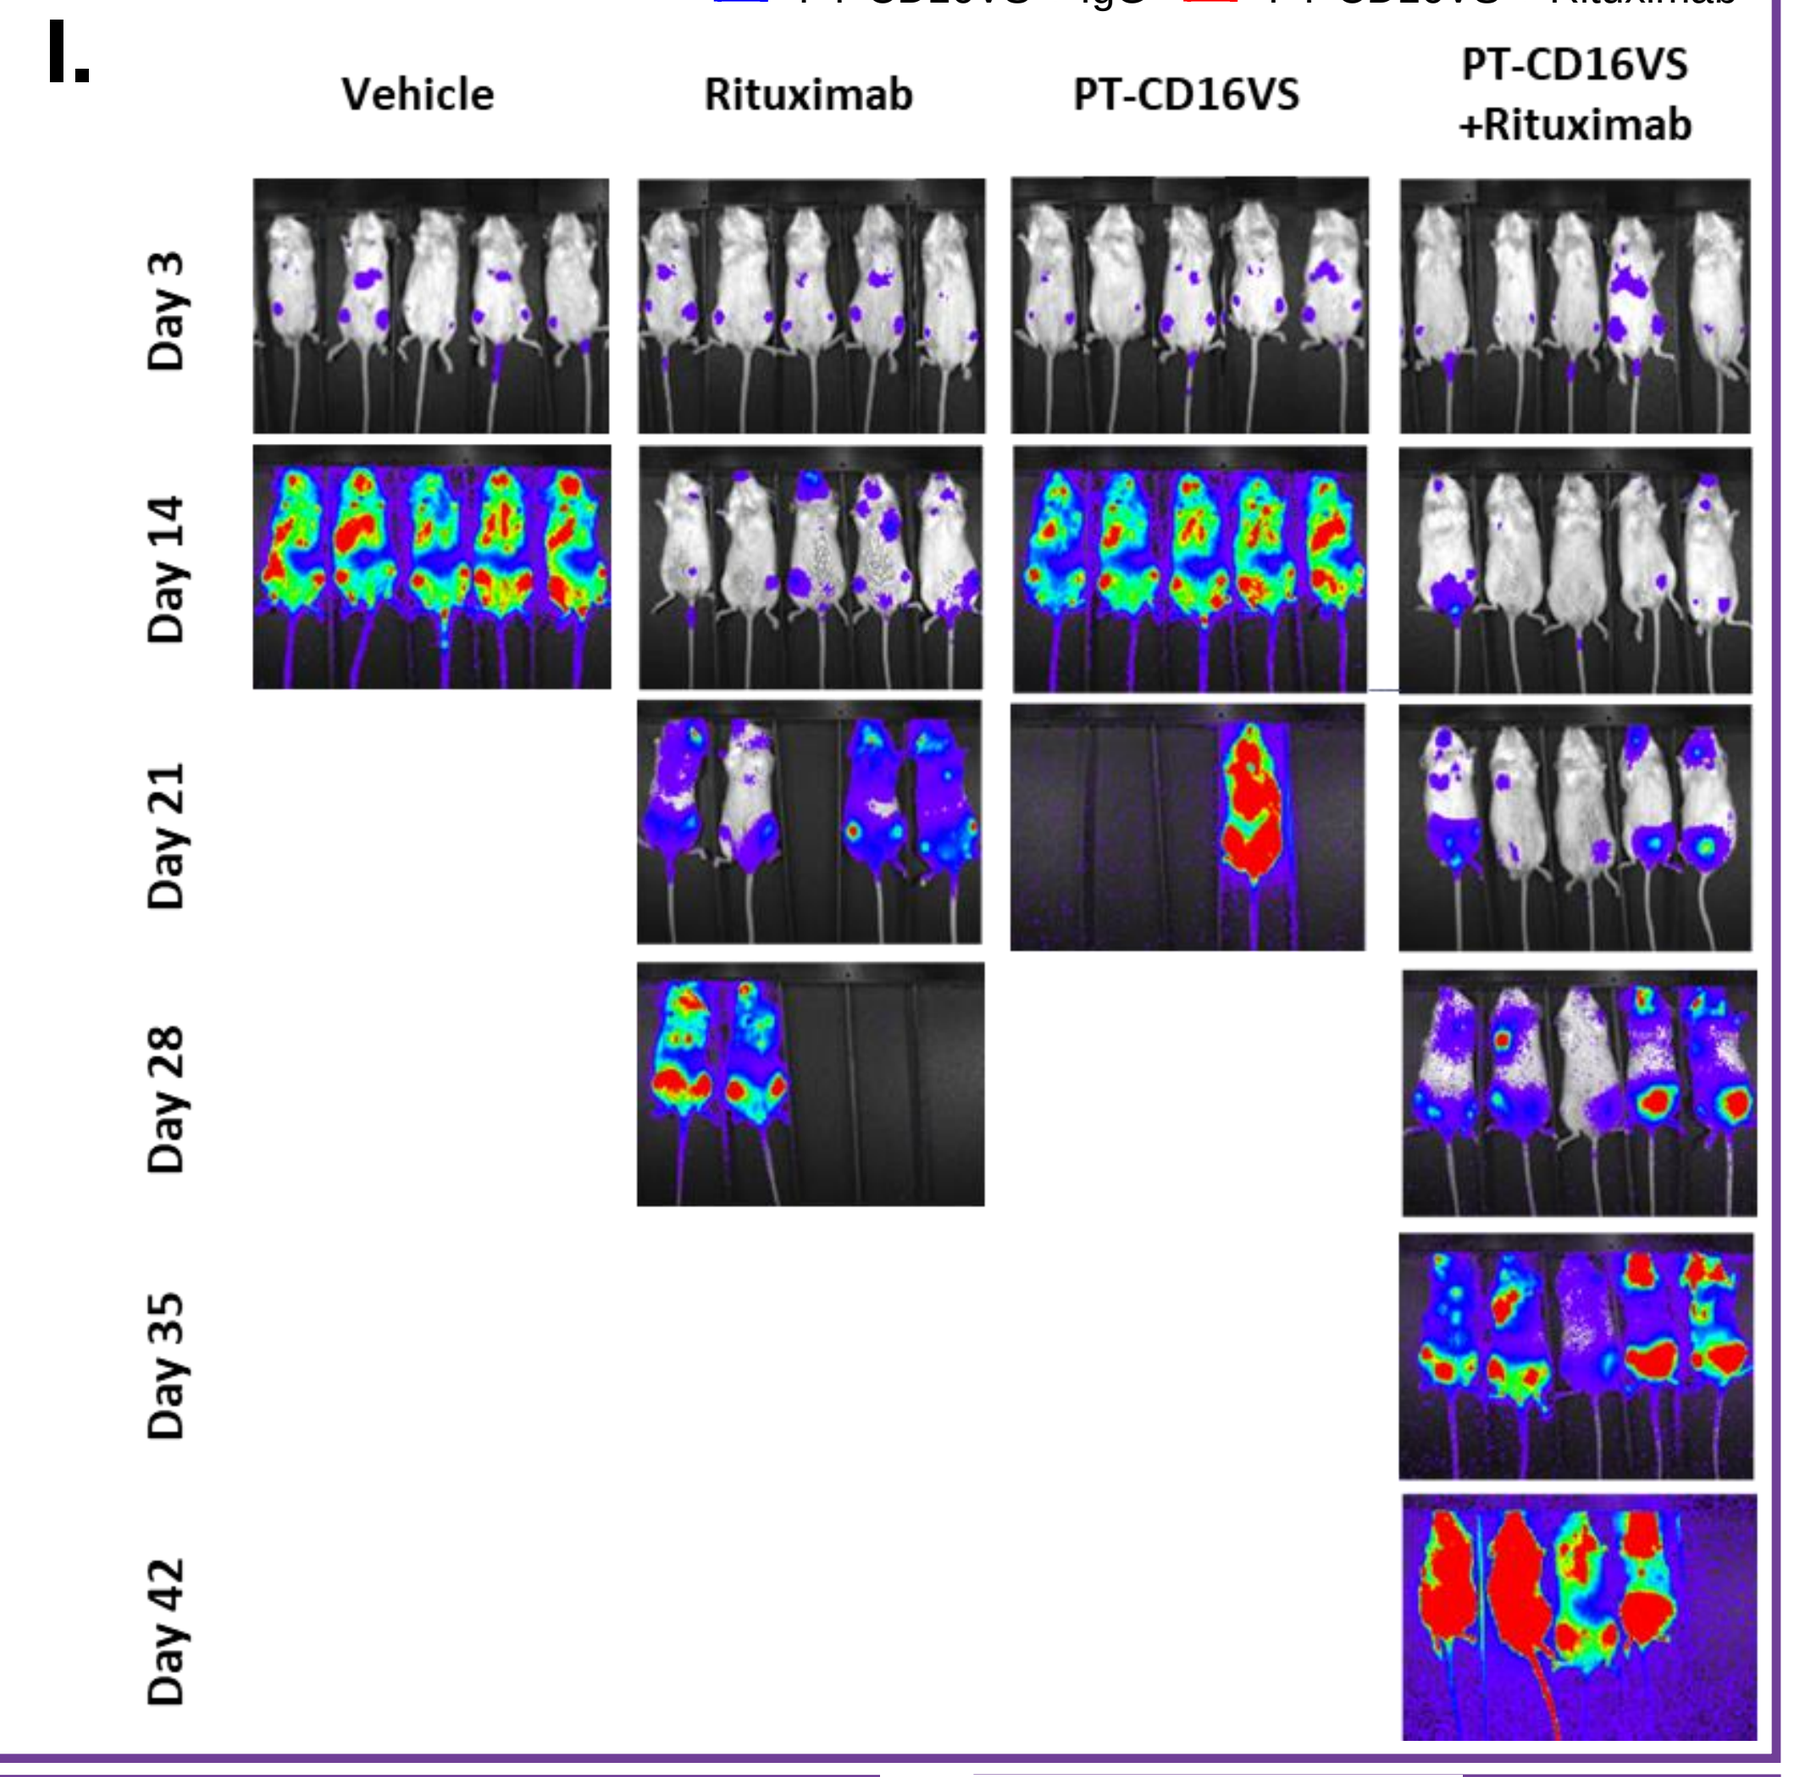
I.
Vehicle
Rituximab
PT-CD16VS
PT-CD16VS +Rituximab
Day 3
Day 14
Day 21
Day 28
Day 35
Day 42

Using A Universal Receptor Approach , A Single Drug Product Can Target Multiple Cancers Across Hematological And Solid Tumors When Combined With Various Monoclonal Antibodies

Corresponding author : Kathy.Karasiewicz@celularity.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.