Developing placental CD34+-derived natural killer cells with high affinity cleavage resistant CD16 (CYNK-101) and Cetuximab for enhanced therapy of EGFR+ non-small cell lung and head and neck cancers

Irene Raitman*, John Fitzgerald*, Valentina Rousseva, Salvatore Rotondo, Xuan Guo, Hemlata Rana, Andrea DiFiglia, Tanel Mahlakõiv, Shuyang He, Lin Kang, Robert Hariri and Xiaokui Zhang

*These authors contributed equally

Celularity Inc., Florham Park, NJ

Abstract #159

INTRODUCTION

Celularity Inc. has developed a novel platform that enables production of a genetically modified allogenic, off-the-shelf, ex vivo expanded and cryopreserved human placental CD34+-derived natural killer (NK) cell therapy named CYNK-101 for the treatment of various cancers.[1] NK cells are key mediators of antibody dependent cellular cytotoxicity (ADCC) via the CD16 receptor on NK cells, which recognizes antibody Fc on the tumor target cells. NK cellular therapies can thus be targeted to tumor antigens when combined with tumor specific antibodies.

CYNK-101 cells express a genetically modified version of the CD16 receptor. The 158Val/Val allele of CD16 has been shown to have higher IgG binding affinity compared to the 158Phe/Phe form,[2] and has been found in ~10-20% of the human population.[3,4] Furthermore, following NK cell activation CD16 on the surface is protease cleaved, which results in decreased ADCC. However, a mutation at Ser197 to Proline has been shown to prevent this cleavage and would thus increase the ADCC activity of NK cells.[5] CYNK-101 described herein express this high IgG affinity and cleavage resistant variant of CD16 (CD16VP) for the purposes of increased ADCC.

The human epidermal growth factor receptor (EGFR) has been found to be overexpressed in 40-90% of non-small cell lung cancers (NSCLC), with squamous cell carcinomas making up the larger proportion, and in as many as 80% of invasive head and neck squamous cell carcinomas (HNSCC); often translating to a worse outcome for patients.[6,7] Cetuximab is a chimeric mouse-human monoclonal antibody that inhibits ligand binding to EGFR. Cetuximab has been shown to inhibit proliferation and increase apoptosis and to induce an ADCC immune cell response when bound to EGFR on the surface of tumor cells.[7,8]

Here we report on the use of CYNK-101, a NK cell immunotherapy product with improved and prolonged anti-tumor activity. CYNK-101 cells were functionally evaluated for their ADCC activity against EGFR+ NSCLC and HNSCC tumor cells in combination with Cetuximab.

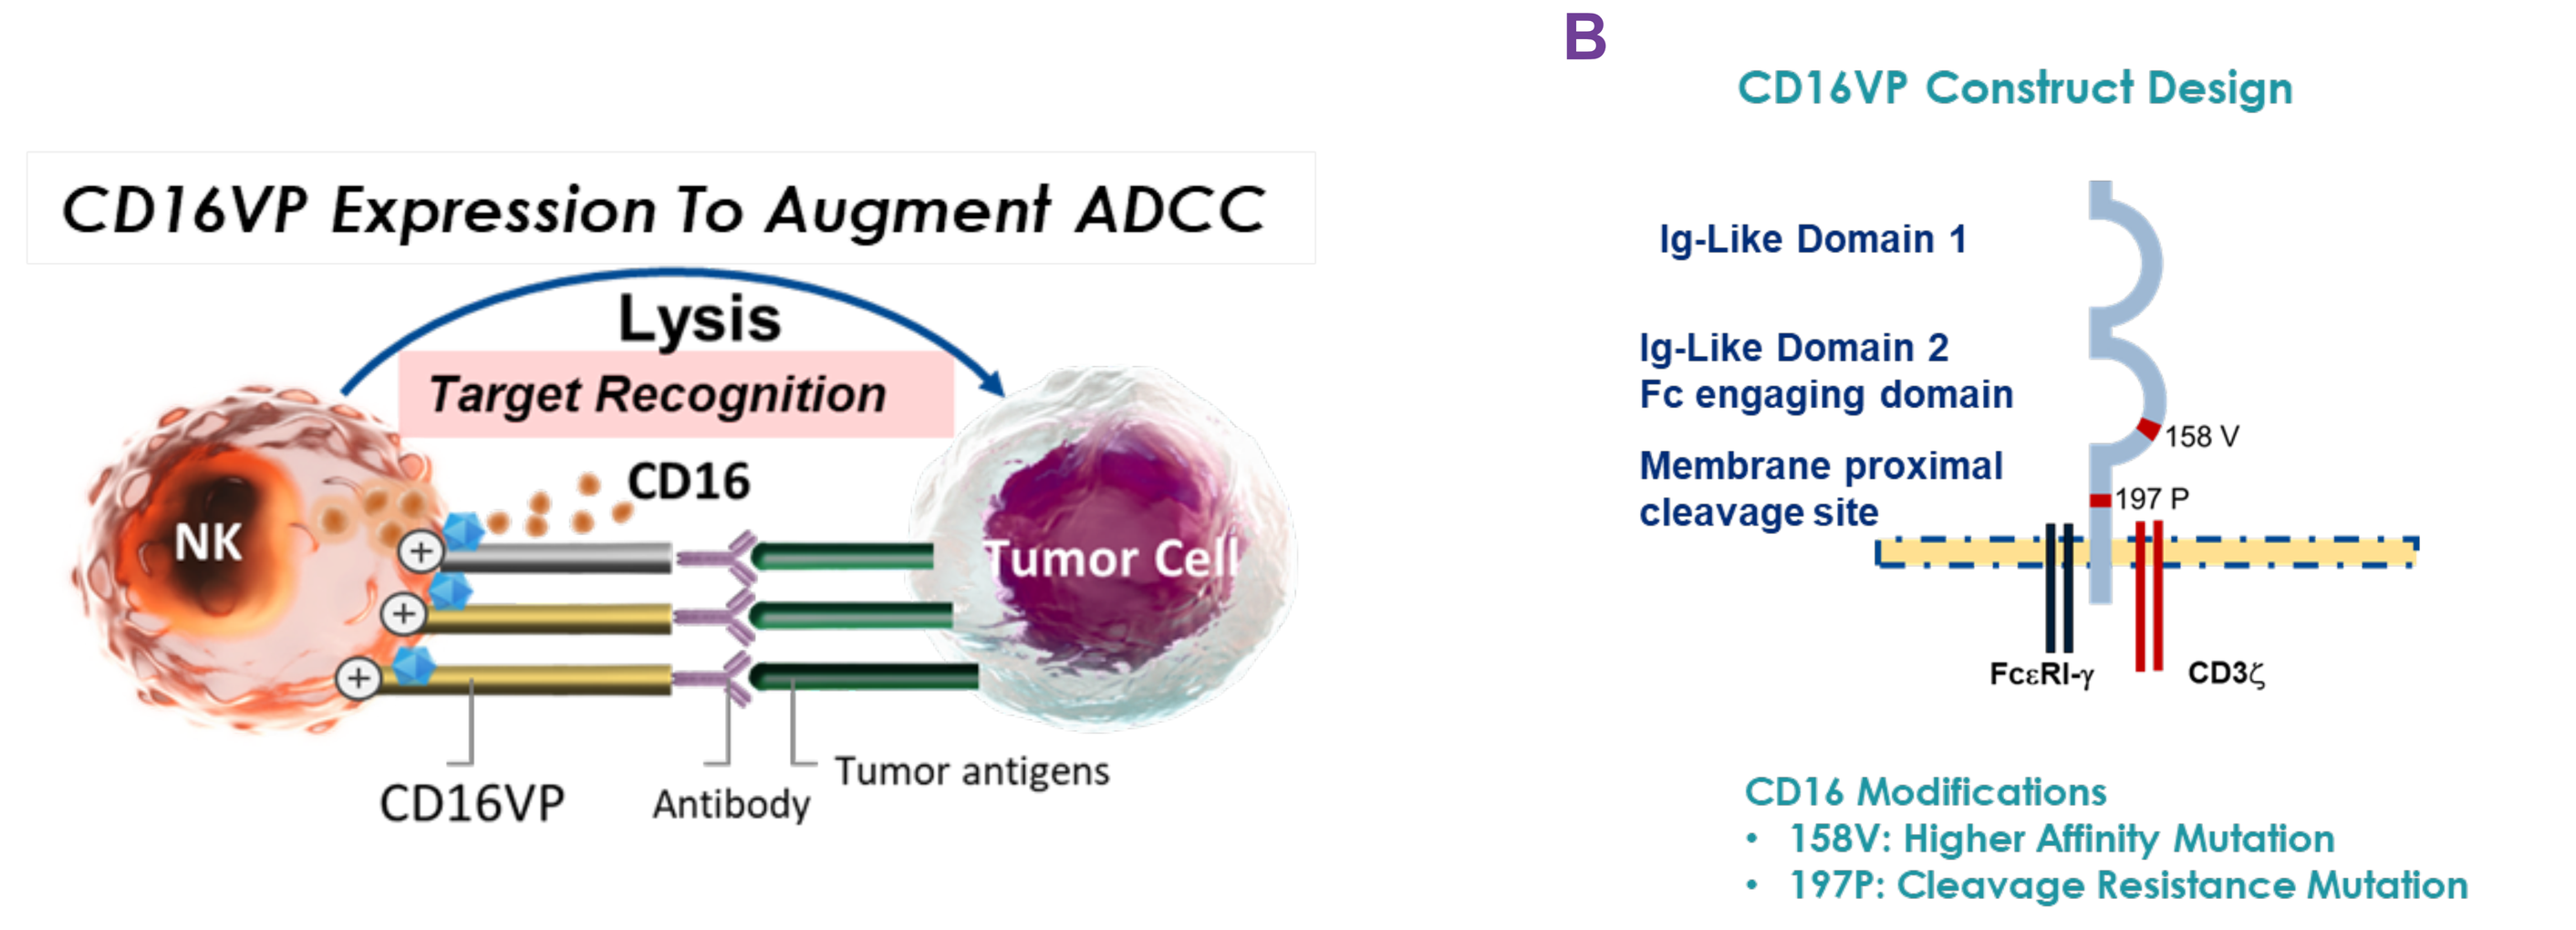
A
CD16VP Expression To Augment ADCC
Lysis
Target Recognition
CD16
NK
Tumor Cell
CD16VP
Antibody
Tumor antigens
B
CD16VP Construct Design
Ig-Like Domain 1
Ig-Like Domain 2
Fc engaging domain
Membrane proximal
cleavage site
158 V
197 P
FcεRI-γ
CD3ζ
CD16 Modifications
• 158V: Higher Affinity Mutation
• 197P: Cleavage Resistance Mutation

Figure 1. (A) Schematic representation of CYNK-101 mediated ADCC. (B) Engineered construct of CD16VP with high affinity and protease cleavage resistant mutations that were expressed on placental CD34+ hematopoietic stem cell derived CYNK-101 cells.

MATERIALS AND METHODS

- Cell culture: Human placental CD34+ cells were transduced with a lentivirus expressing a high binding affinity (158Val) and protease resistant (197Pro) CD16 variant (CD16VP) and cultured in the presence of cytokines to generate CYNK-101 cells.
- Cell characterization: Upon completion of cell expansion and differentiation CYNK-101 cells were frozen. All subsequent characterization and functional analyses were performed on the frozen product after thaw. CYNK-101 cells were evaluated for NK surface markers CD56, CD3, and CD16 by flow cytometry immediately post thaw.
- Evaluation of CYNK-101 anti-tumor activity in combination with Cetuximab: The anti-tumor activity of CYNK-101 cells against EGFR-positive NSCLC cell lines SK-MES-1 and NCI-H226 and HNSCC cell lines CAL-27, FaDu, A-253, and SCC-25 was assessed in combination with 0.1 µg/ml Cetuximab or the IgG control antibody at various effector to target (E:T) ratios. ADCC activity was measured by real-time xCELLigence (ACEA Biosciences) and cytokine secretion by Luminex xMAP multiplex assay (Millipore Sigma). The phosphatidylinositol 3-kinase (PI3K) inhibitor Wortmannin was used to study the molecular mechanisms underlying cytotoxicity of CYNK-101 cells against NSCLC and HNSCC.
- Statistical Analysis: Statistical analysis was performed using GraphPad Prism and Excel programs. Data are presented as mean ± standard deviation. Paired 2-tailed Student's t-tests were used for comparing 2 groups.

SUMMARY

- Celularity has developed genetically modified human placental CD34+-derived, cryopreserved, off-the-shelf, allogenic NK cells (CYNK-101) with a high IgG binding affinity and proteinase resistant CD16 variant (CD16VP) for cancer treatment.
 - >95% NK purity was achieved by the process
 - ~72% or greater CD16VP expression was maintained post thaw, and the CD16 was resistant to being cleaved from the surface following NK cell activation
- CYNK-101 cells demonstrated enhanced Cetuximab-mediated ADCC activity against EGFR+ NSCLC and HNSCC tumor cell lines.
- CYNK-101, when in combination with Cetuximab, secreted higher levels of GM-CSF, IFN-γ, and TNF-α against A-253, FaDu, and SCC-25 HNSCC cell lines, and increased GM-CSF and IFN-γ against the NSCLC cell line NCI-H226.
- The Cetuximab mediated increased cytotoxicity was PI3K pathway-dependent as Wortmannin decreased the ADCC.
- CYNK-101 cells provide a combination immunotherapy option by harnessing the anti-tumor activities of both targeted biologics and innate cytotoxicity of NK cells. Further development of CYNK-101 in combination with the therapeutic antibody for these solid tumor indications is warranted.

RESULTS

Figure 2. EGFR Expression of NSCLC and HNSCC Cell Lines

A

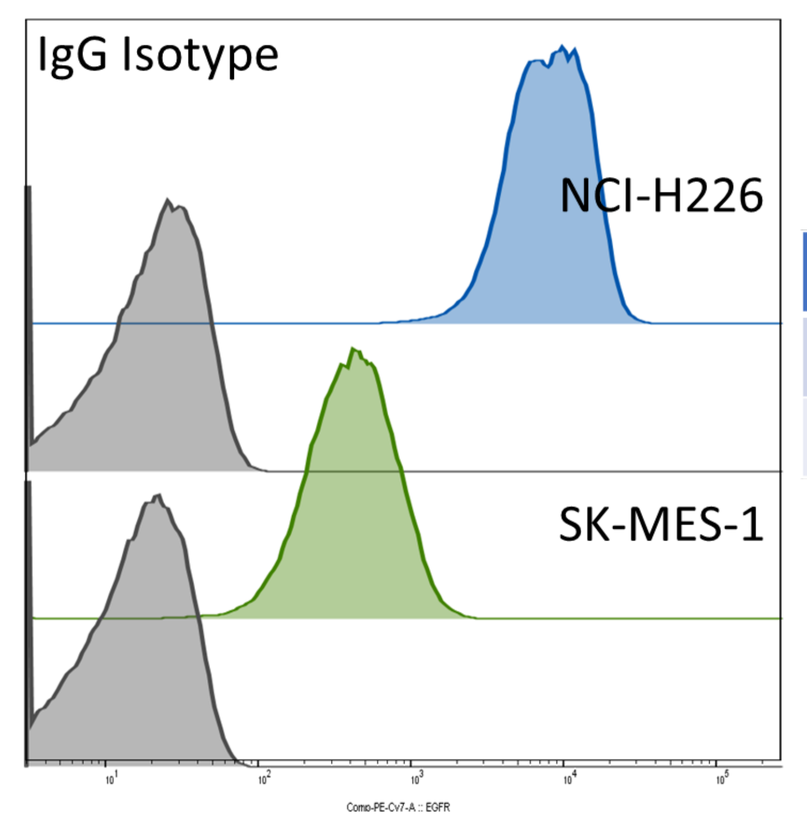
IgG Isotype
NCI-H226
SK-MES-1

Cell Line	EGFR+ (%)	EGFR MFI
NCI-H226	100.0	7703
SK-MES-1	99.0	408

B

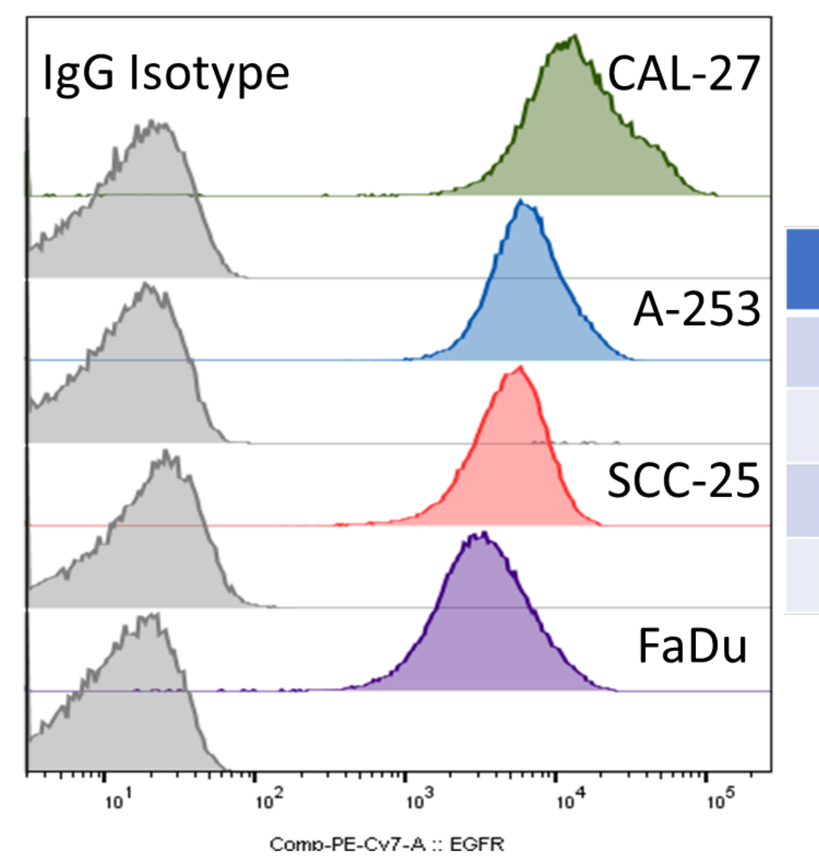
IgG Isotype
CAL-27
A-253
SCC-25
FaDu
Comp-PE-Cy7-A :: EGFR

Cell Line	EGFR+ (%)	EGFR MFI
CAL-27	98.4	13398
A-253	99.7	6358
SCC-25	99.7	4574
FaDu	99.5	3203

Figure 2. EGFR expression in NSCLC (A) and HNSCC (B) tumor cell lines. Isotype-stained cells, shown in grey, were used as gating controls for each cell line. The percentage of tumor cells expressing EGFR and the geometric mean fluorescence intensity (MFI) for each of the cell lines are indicated in the adjacent tables.

Figure 3. High Expression and Cleavage Resistance of CD16 on CYNK-101 Cells

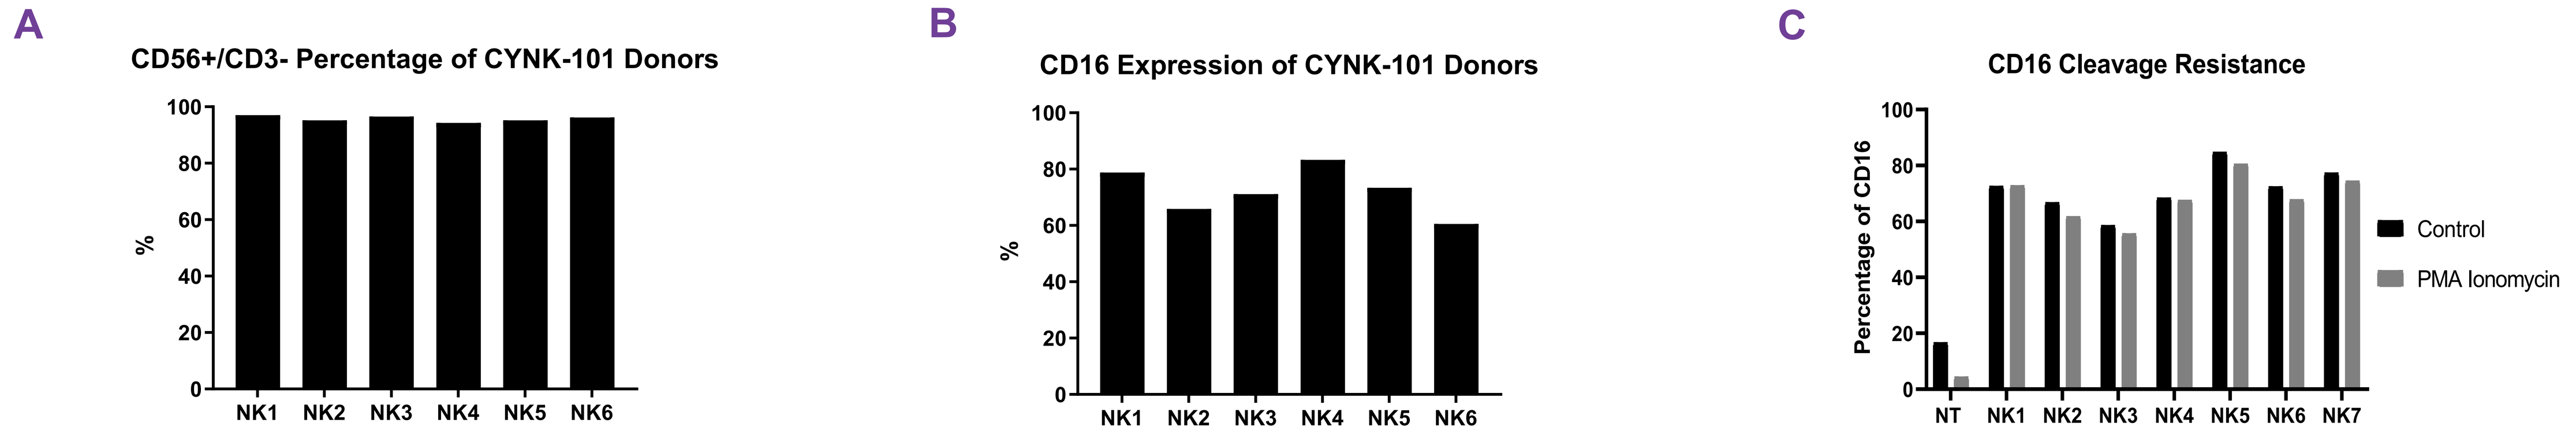
A
CD56+/CD3- Percentage of CYNK-101 Donors
B
CD16 Expression of CYNK-101 Donors
C
CD16 Cleavage Resistance
Percentage of CD16
Control
PMA Ionomycin
NT NK1 NK2 NK3 NK4 NK5 NK6 NK7

Figure 3. (A) Percentage of cells CD56+/CD3- in CYNK-101 donors post thaw, 95.7% ± 1.0% (mean ± SD) for the 6 donors shown. (B) CD16 expression of CD56+/CD3- CYNK-101 cells post thaw, 72.2% ± 8.3% (mean ± SD) for the 6 donors shown. (C) Quantification of CD16 cleavage resistance, shown via percentage of CD16 on the surface of CD56+/CD3- cells, in CYNK-101 (mean ± SD, n=7 donors) and lack of cleavage resistance in a NT (non-transduced) donor following 4h activation by PMA/ionomycin.

Figure 4. CYNK-101 Cells in Combination with Cetuximab Have Increased ADCC Against NSCLC and HNSCC Cell Lines

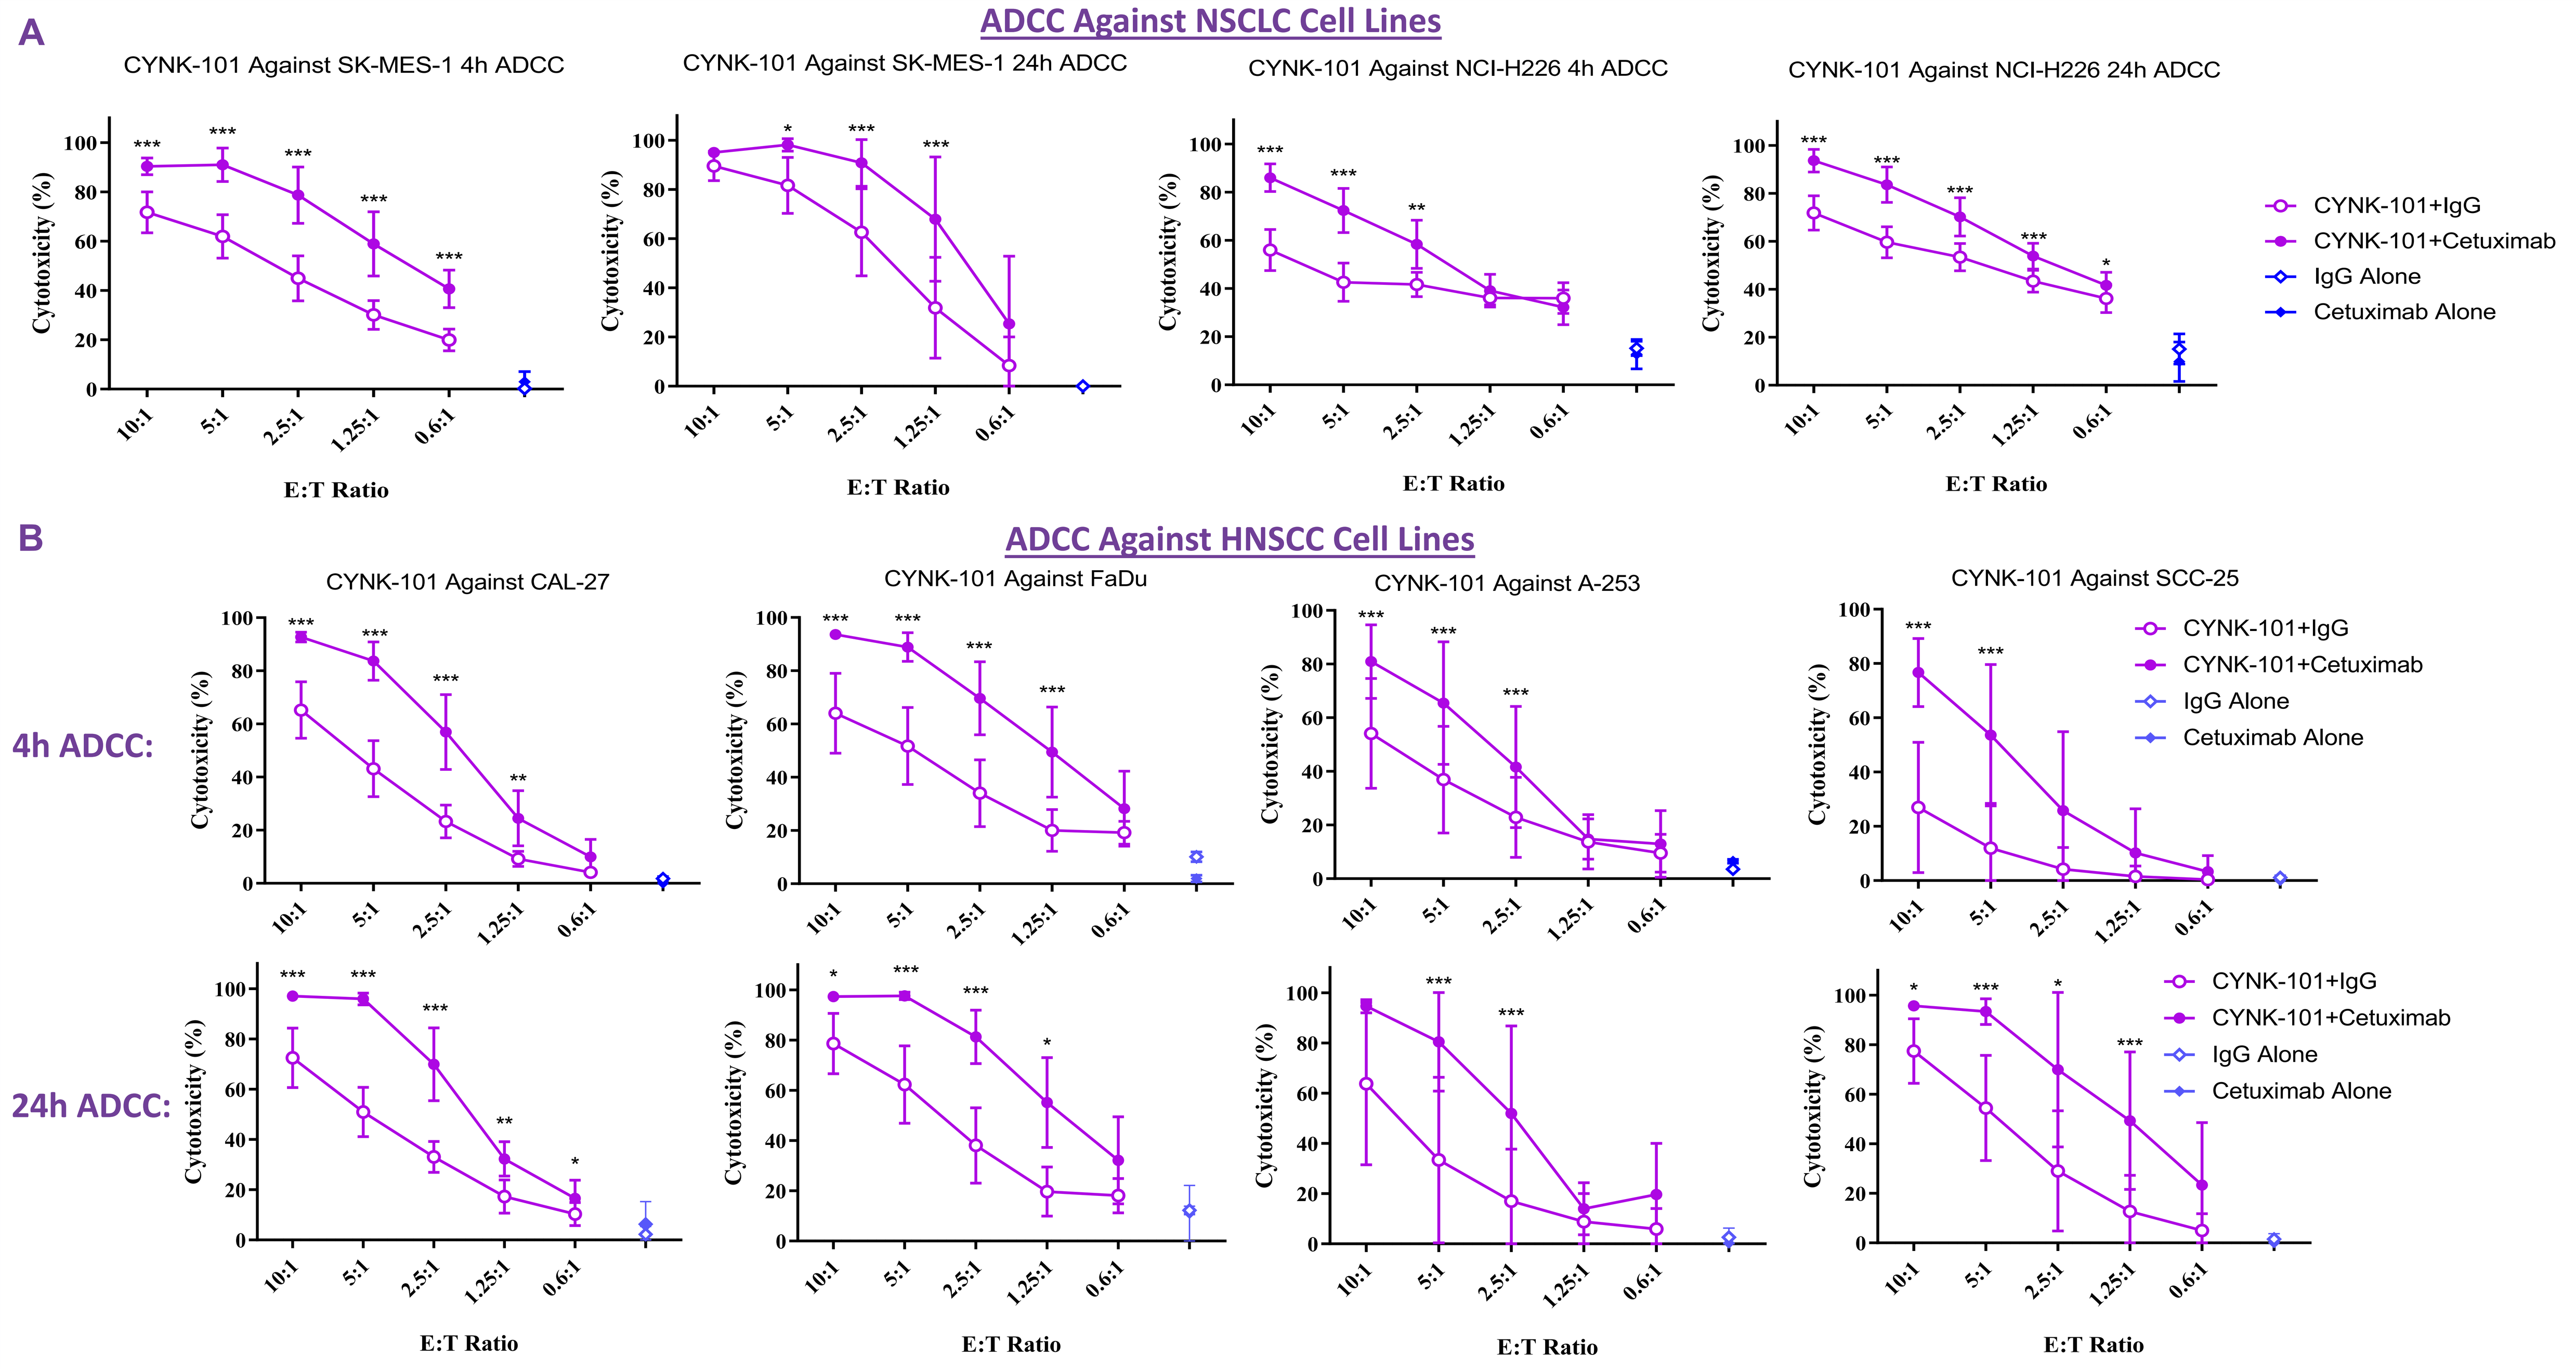
A
ADCC Against NSCLC Cell Lines
CYNK-101 Against SK-MES-1 4h ADCC
CYNK-101 Against SK-MES-1 24h ADCC
CYNK-101 Against NCI-H226 4h ADCC
CYNK-101 Against NCI-H226 24h ADCC
Cytotoxicity (%)
E:T Ratio
CYNK-101+IgG
CYNK-101+Cetuximab
IgG Alone
Cetuximab Alone
B
ADCC Against HNSCC Cell Lines
CYNK-101 Against CAL-27
CYNK-101 Against FaDu
CYNK-101 Against A-253
CYNK-101 Against SCC-25
4h ADCC:
24h ADCC:

Figure 4. 4h and 24h ADCC activity (mean ± SD) of CYNK-101 cells in combination with Cetuximab compared to the control IgG antibody against NSCLC cell lines SK-MES-1 and NCI-H226 (A, n=5 donors) and HNSCC cell lines CAL-27, FaDu, A-253, and SCC-25 (B, n=6 donors) at the indicated E:T ratios. Cytotoxicity from Cetuximab or IgG alone is included for reference. * indicate significantly higher activity with Cetuximab compared to IgG (***p<0.005, **p<0.01, and *p<0.05).

Figure 5. CYNK-101 in Combination with Cetuximab Results in Increased Cytokine Secretion Against NSCLC and HNSCC Cell Lines

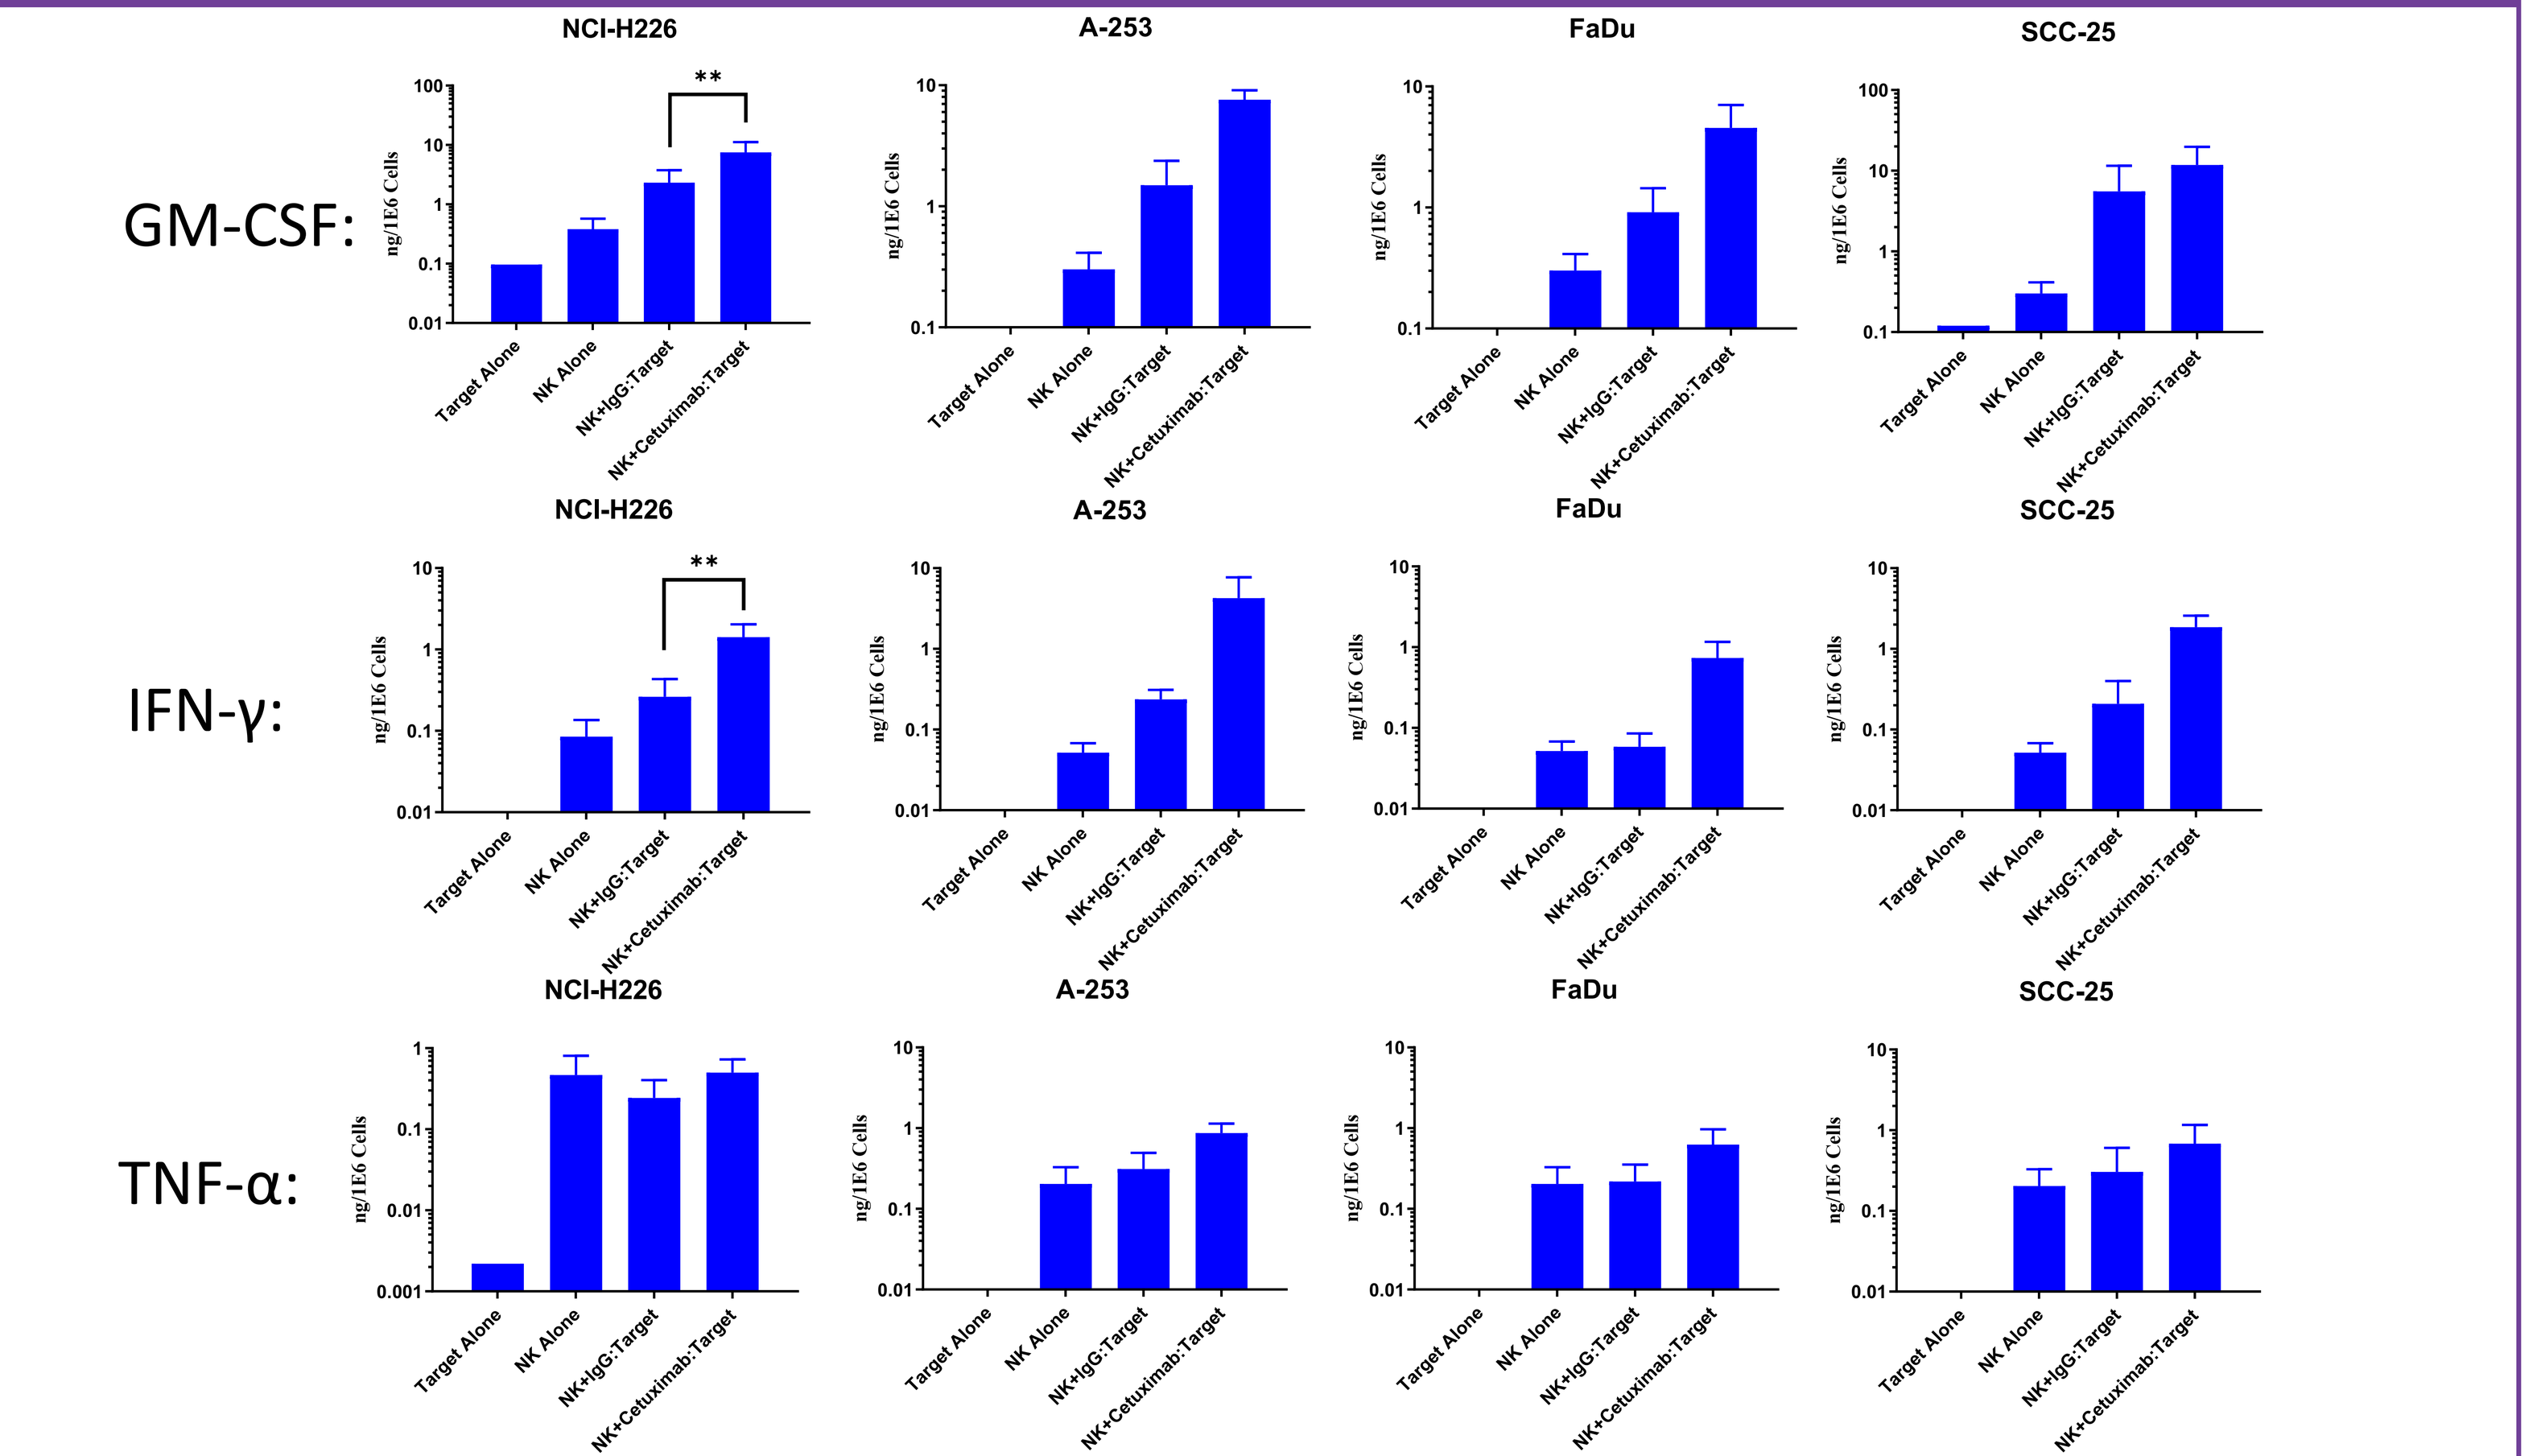
GM-CSF:
IFN-γ:
TNF-α:
NCI-H226
A-253
FaDu
SCC-25
ng/1E6 Cells
Target Alone
NK Alone
NK+IgG:Target
NK+Cetuximab:Target

Figure 5. Level of the inflammatory cytokines GM-CSF, IFN-γ, and TNF-α (ng/1E6 cells) in culture supernatants following 24h co-culture at a 1:1 E:T ratio of CYNK-101 cells in combination with Cetuximab or IgG control and the NSCLC cell line NCI-H226 or the HNSCC cell lines A-253, FaDu, or SCC-25 (mean ± SD, n=6 donors (n=5 for NCI-H226)). The cytokine secretion from target cells without the presence of CYNK-101 cells is also included for reference, as well as that from CYNK-101 cells alone. * indicate significantly higher secretion of the indicated cytokine from CYNK-101 cells when in combination with Cetuximab compared to IgG control (*p<0.05, **p<0.01, and ***p<0.005).

Figure 6. Increased ADCC of CYNK-101 in Combination with Cetuximab is PI3K Pathway Dependent

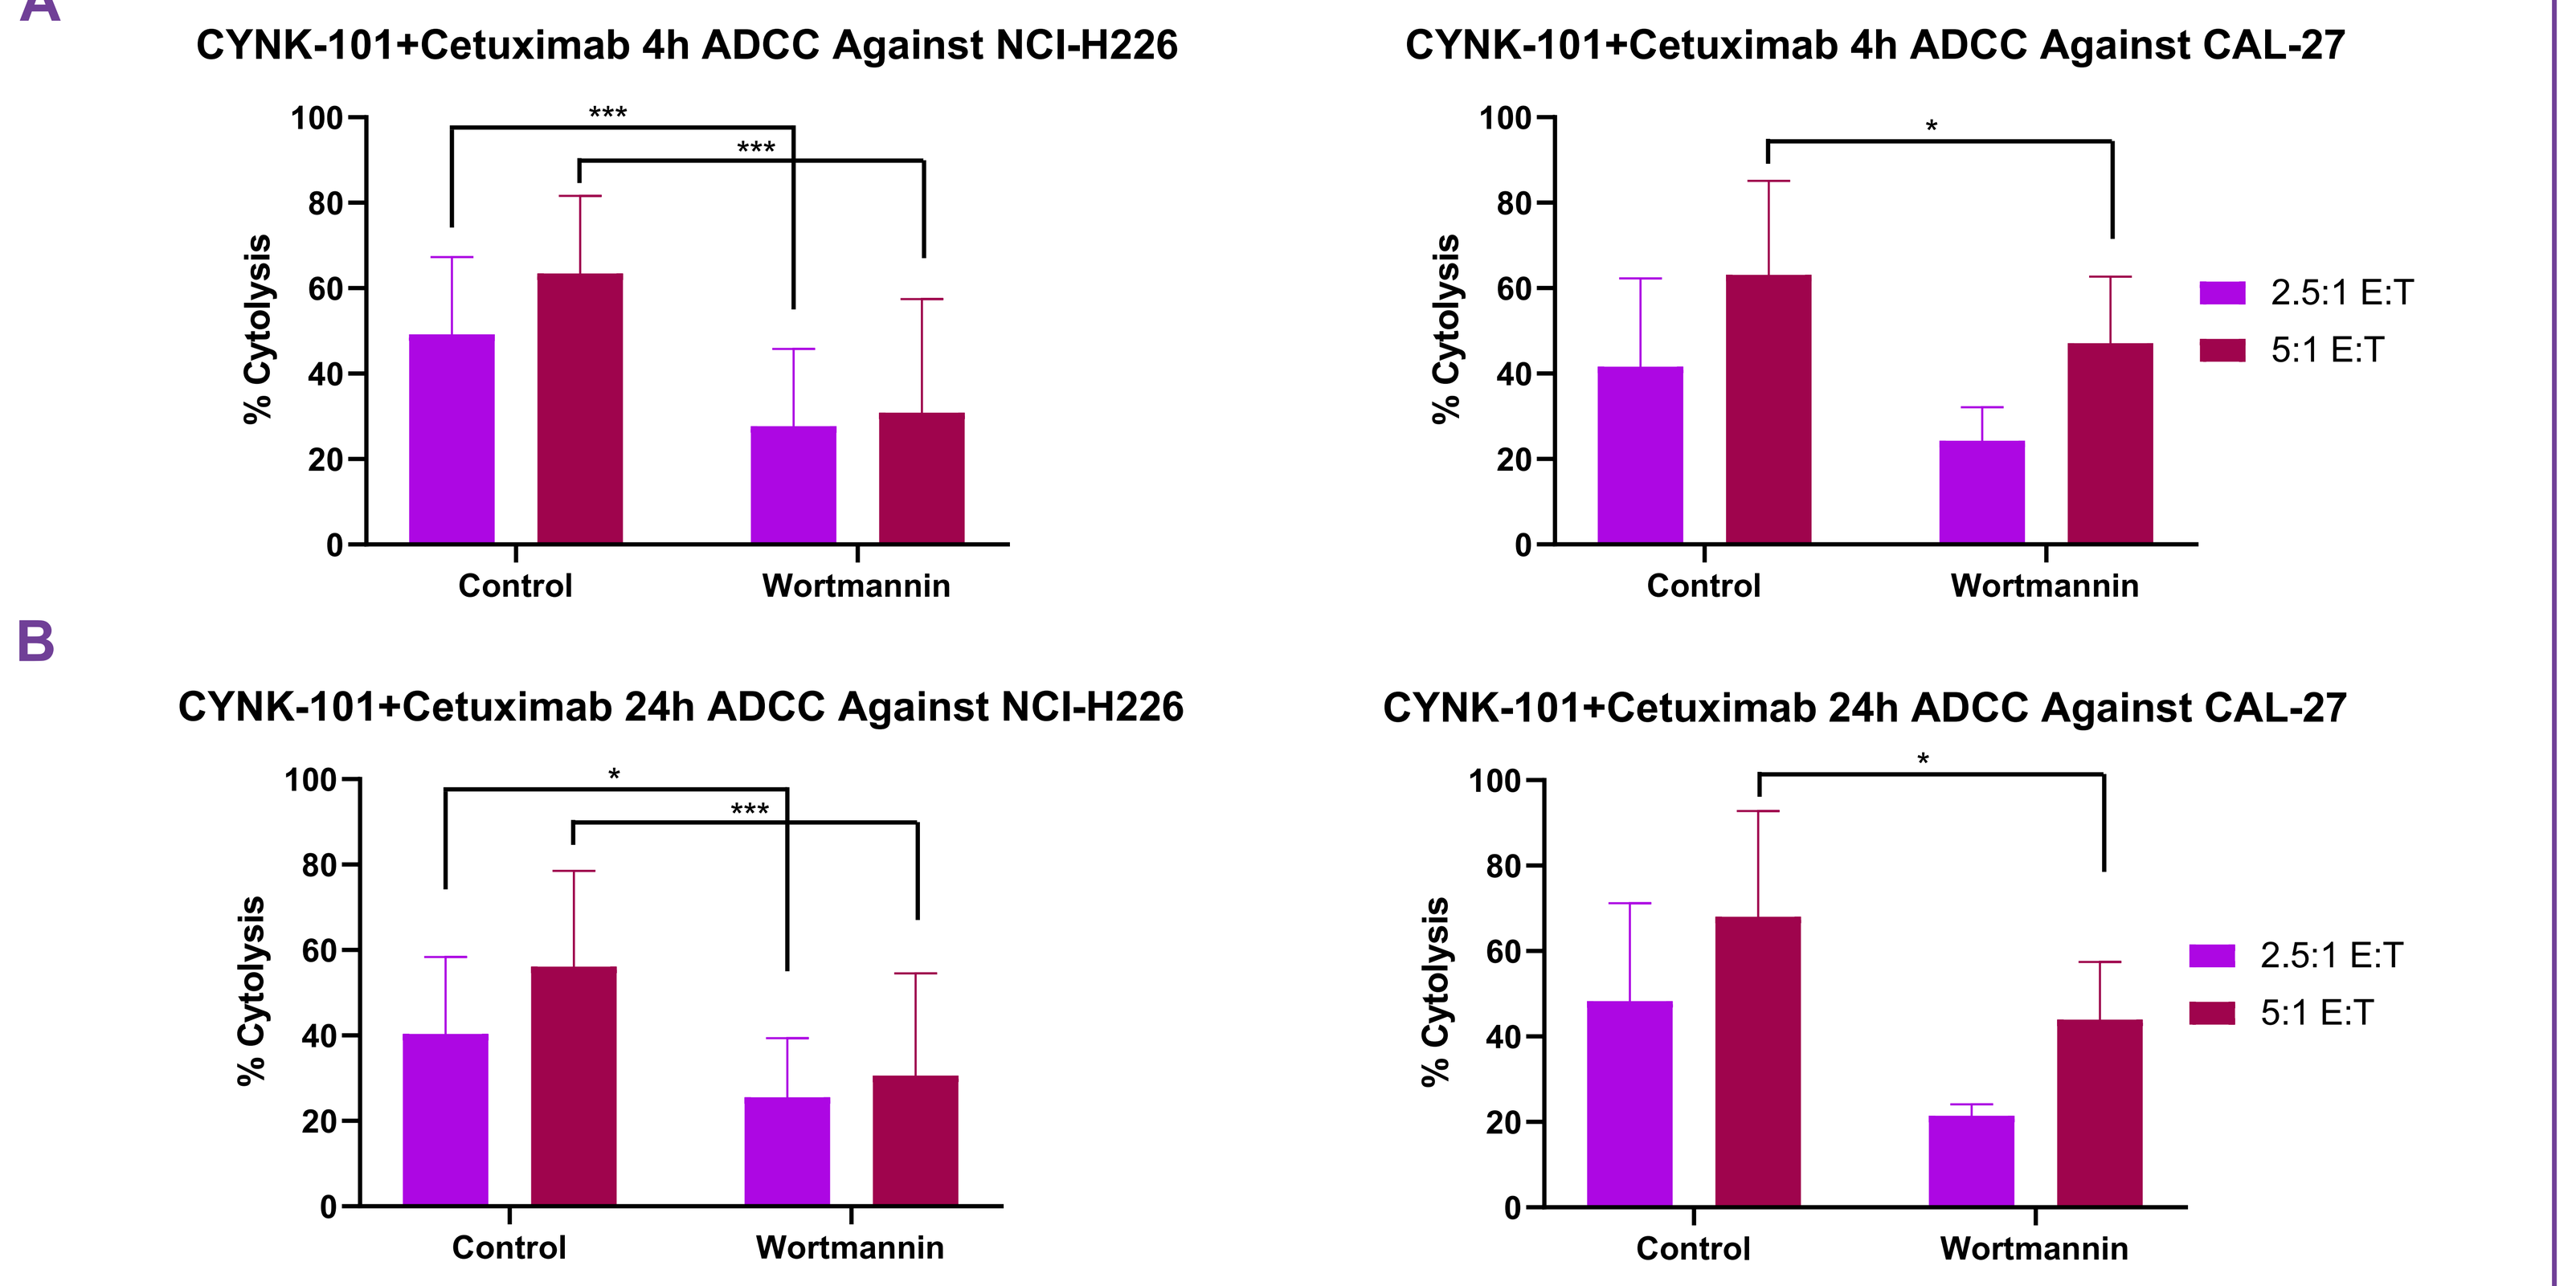
A
CYNK-101+Cetuximab 4h ADCC Against NCI-H226
CYNK-101+Cetuximab 4h ADCC Against CAL-27
B
CYNK-101+Cetuximab 24h ADCC Against NCI-H226
CYNK-101+Cetuximab 24h ADCC Against CAL-27
% Cytolysis
Control
Wortmannin
2.5:1 E:T
5:1 E:T

Figure 6. 4h (A) and 24h (B) cytolysis of untreated or 1h 0.1 µM Wortmannin pretreated CYNK-101 cells in combination with Cetuximab against the NSCLC cell line NCI-H226 (left) and the HNSCC cell line CAL-27 (right) at the 2 indicated E:T ratios (mean ± SD, n=4 donors) * indicate significantly decreased cytolysis with the PI3K inhibitor compared to control at that E:T ratio (*p<0.05 and ***p<0.005).

References

1. Kang L, et al. Front Immunol. 2013;4: 101.
2. Wu J et al. J Clin Invest. 1997;100(5):1059-1070.
3. Sugita N et al. Clin Exp Immunol. 1999;117(2):350-354.
4. Koene HR et al. Blood. 1997;90(3):1109-1114.
5. Jing Y et al. PLoS One. 2015;10(3):e0121788.
6. Prabhakar C. Transl Lung Cancer Res. 2015;4(2): 110-118.
7. Ratushny V et al. Cell Signal. 2009;21(8): 1255–1268.
8. Kurai J et al. Clin Cancer Res. 2007;13(5): 1552-1561.

Disclosure

IR: JF: VR: SR: XG: HR: AD: TM: SH: LK: RH: XZ: Celularity Inc. Employment